DocuSign Envelope ID: 3CC26EEA-F429-483F-824B-79B644D57661

AASLD Nov. 4-8, 2022 The Liver Meeting®

Nonclinical efficacy, pharmacokinetic profile and pharmacokinetic/pharmacodynamic (PK/PD) correlation of ALG-125755, a GalNAc-conjugated siRNA, for functional cure of chronic hepatitis B

Kusum Gupta[1], Megan Fitzgerald[1], Kha Le[1], Vikrant Gohil[1], Sucheta Mukherjee[1], Dinah Misner[1], Jin Hong[1], Hyunsoon Kang[1], Saul Martinez Montero[1]; Vivek K. Rajwanshi[1], Felix Lai[1], Meenakshi Venkatraman[1], Mathew McClure[1], Sushmita Chanda[1], John Fry[1], David B. Smith[1], Julian A. Symons[1], Lawrence M. Blatt[1], Leonid N. Beigelman[1], and Tse-I Lin[2]

[1]Aligos Therapeutics, Inc., South San Francisco, CA; [2]Aligos Belgium BV, Leuven, Belgium

Email: kgupta@aligos.com

Publication Number: 1155

Background and Aim

Chronic hepatitis B (CHB) is a major global health problem. The high morbidity and mortality associated with the disease emphasizes the need for a functional cure of CHB. We are developing ALG-125755, a N-acetylgalactosamine (GalNAc)-conjugated small interfering RNA (siRNA), with the aim of achieving sustained clearance of HBV surface antigen (HBsAg).

Methods

ALG-125755 and its metabolite, ALG-126144 (AS(N-1)3' ALG-127555), were tested in HepG2.2.15 cells infected with HBV Genotype D. PK parameters were obtained following a single short IV infusion at 1.5 mg/kg in monkey, a single subcutaneous (SC) dose at 5 to 15 mg/kg, or 6 weekly SC doses at 10 to 100 mg/kg/dose in rat and monkey. Compound metabolism was evaluated in vitro in plasma and mouse liver homogenate, and in vivo in rat and monkey plasma and tissues. Samples were analyzed by liquid chromatography (LC)-fluorescence detection or LC-mass spectrometry. PK/PD correlation was performed from a previously reported adeno-associated virus (AAV)-HBV mouse efficacy study where 10 mg/kg single dose, 5 mg/kg every other week or every four weeks demonstrated significant and durable decline in serum HBsAg (Fitzgerald et al Poster number SAT 386, EASL 2022). Steady state efficacious human liver concentrations were projected using a population PK model on the data from the in vivo efficacy study.

In vitro Activity of ALG-125755, ALG-125903 (Unconjugated ALG-125755) and ALG-126179 (Unconjugated Metabolite ALG-126144)

Figure 1. ALG-125755, ALG-125903 and ALG-126179 inhibition of HBsAg secretion in HepG2.2.15 Cells

ALG-125755 **ALG-125903** **ALG-126179**

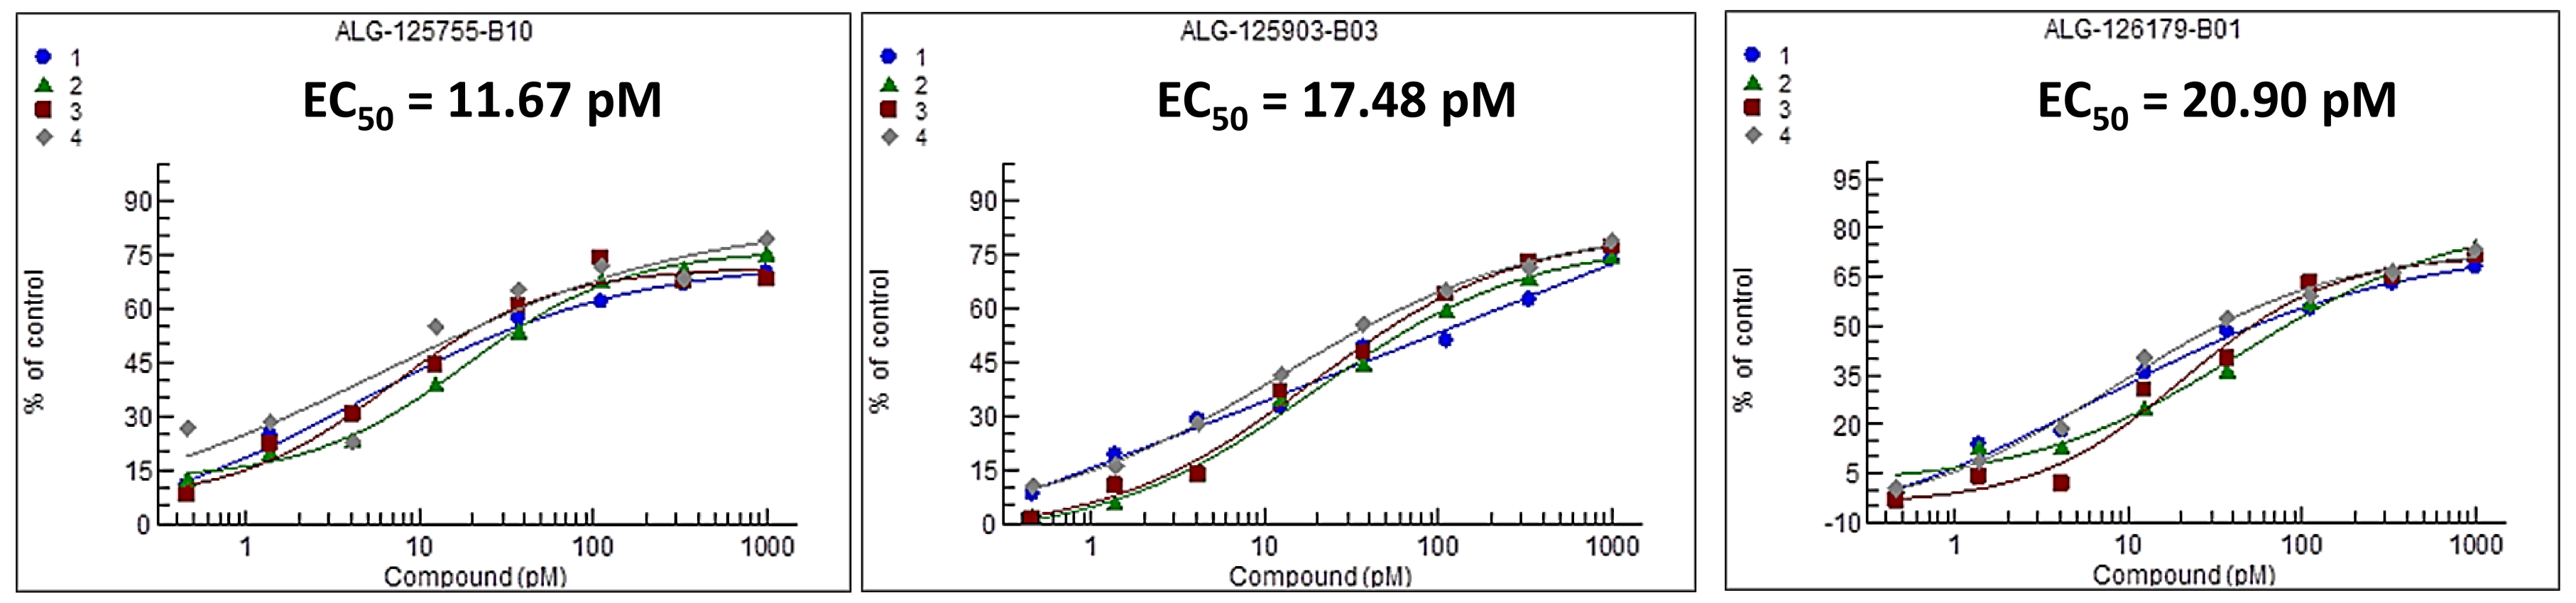
ALG-125755-B10
EC_{50} = 11.67 pM
ALG-125903-B03
EC_{50} = 17.48 pM
ALG-126179-B01
EC_{50} = 20.90 pM
% of control
Compound (pM)

In Vitro EC_{50} and CC_{50}

- **ALG-125755 and its unconjugated form ALG-125903 had comparable EC_{50} values in the HepG2.2.15 cell line**
- **ALG-126179, the unconjugated form of metabolite ALG-126144 (AS(N-1)3' ALG-125755), demonstrated similar in vitro potency to the parent full length siRNA**
- **No cytotoxicity was observed (CC_{50} >1000 pM for all siRNAs)**

Pharmacokinetics Following a Single SC 5 mg/kg Dose in Rat and Monkey

Figure 2. Plasma and liver profile of ALG-125755 in rat and monkey

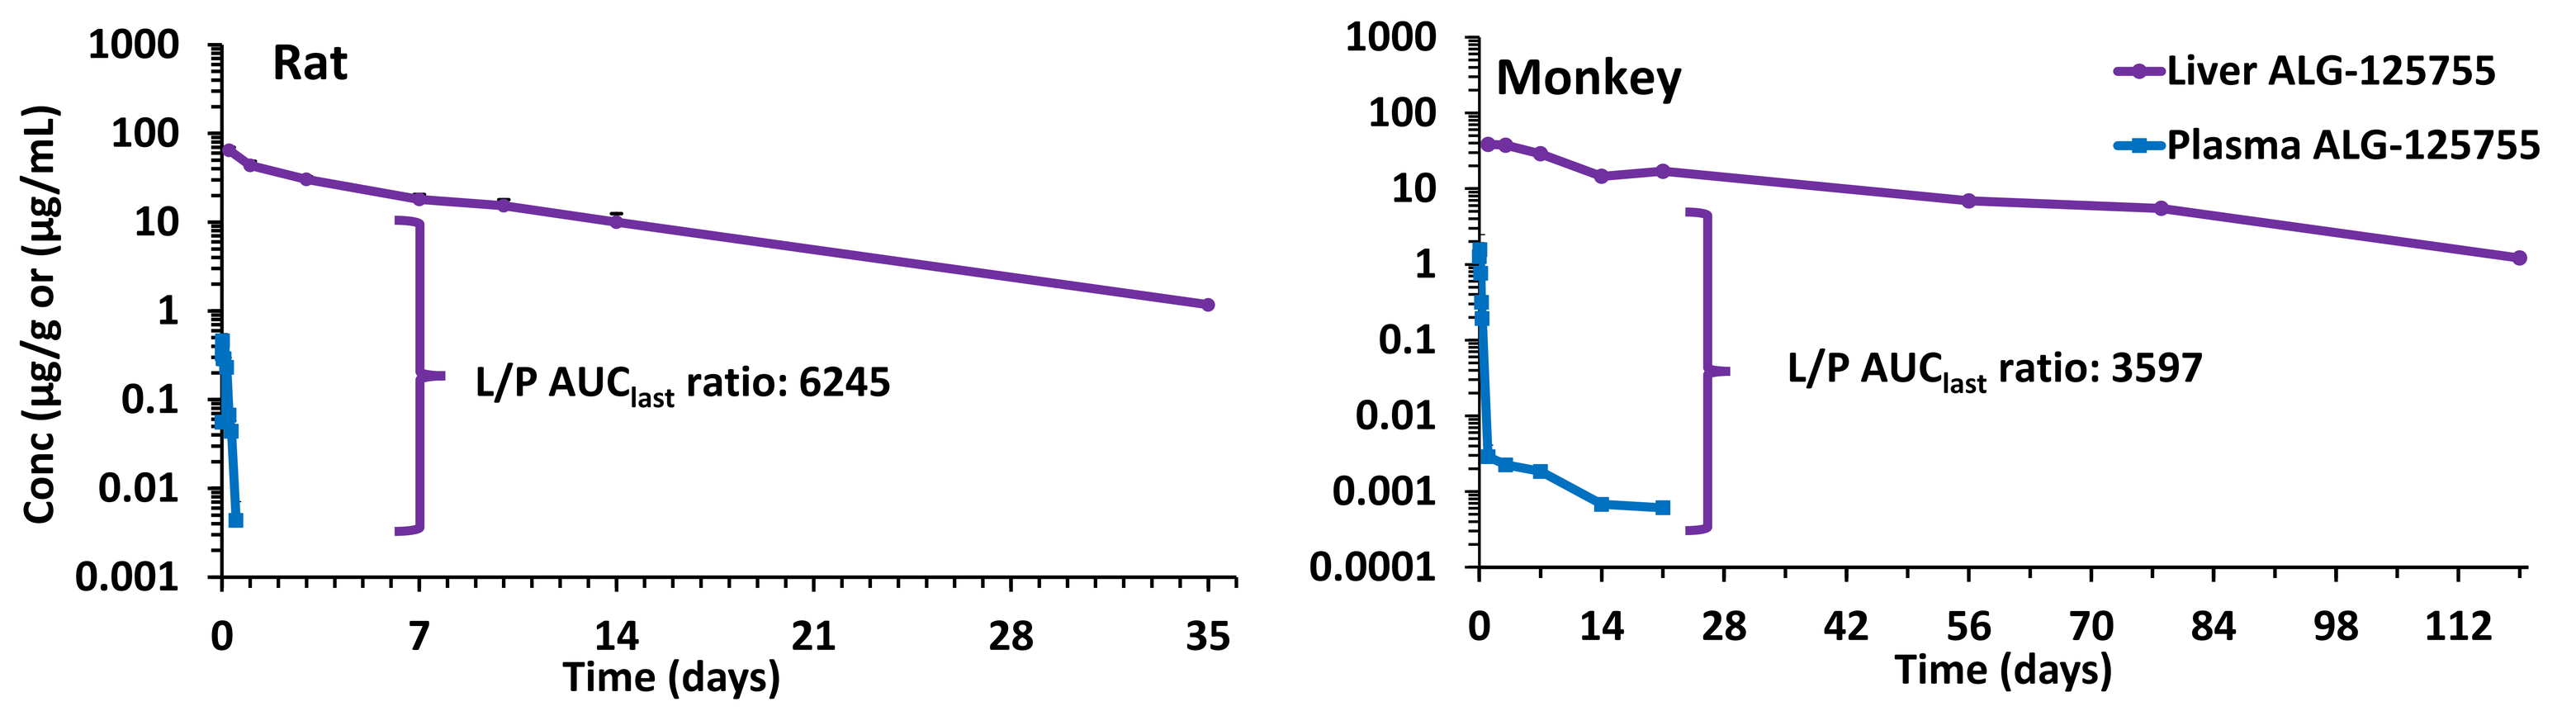
Rat
Monkey
Liver ALG-125755
Plasma ALG-125755
L/P AUC_{last} ratio: 6245
L/P AUC_{last} ratio: 3597
Conc (µg/g or (µg/mL)
Time (days)

L/P = Liver to plasma ratio

Table 1. Plasma and liver PK parameters of ALG-125755 in rat and monkey

Matrix	Rat			Monkey			
	C_{max} (µg/mL or µg/g)	AUC_{last} (µg·h/mL or µg·h/g)	$t_{1/2}$	C_{max} (µg/mL or µg/g)	AUC_{last} (µg·h/mL or µg·h/g)	$t_{1/2}$	F%[a]
Plasma	0.46	1.57	1.4 h	1.56	7.53	2.6 h	62.4
Liver	64.8	9807	6.8 d	38.4	27077	26.4 d	

[a]From AUC_{inf} following IV-infusion for 1 h at 1.5 mg/kg; h=hour, d=day; Plasma analysis by PNA hybridization method, Tissue analysis by LC-HRAM

Single SC Dose PK at 5 mg/kg

- **Rapid absorption and distribution with short plasma half-life**
- **Good systemic bioavailability in monkey**
- **High liver to plasma ratio of ALG-125755**
- **Long half-life in liver supports at least monthly regimen in human**

Liver Concentration Following Repeated SC Doses for 5 Weeks in Rat and Monkey

Figure 3. Liver concentrations of ALG-125755 in rat and monkey

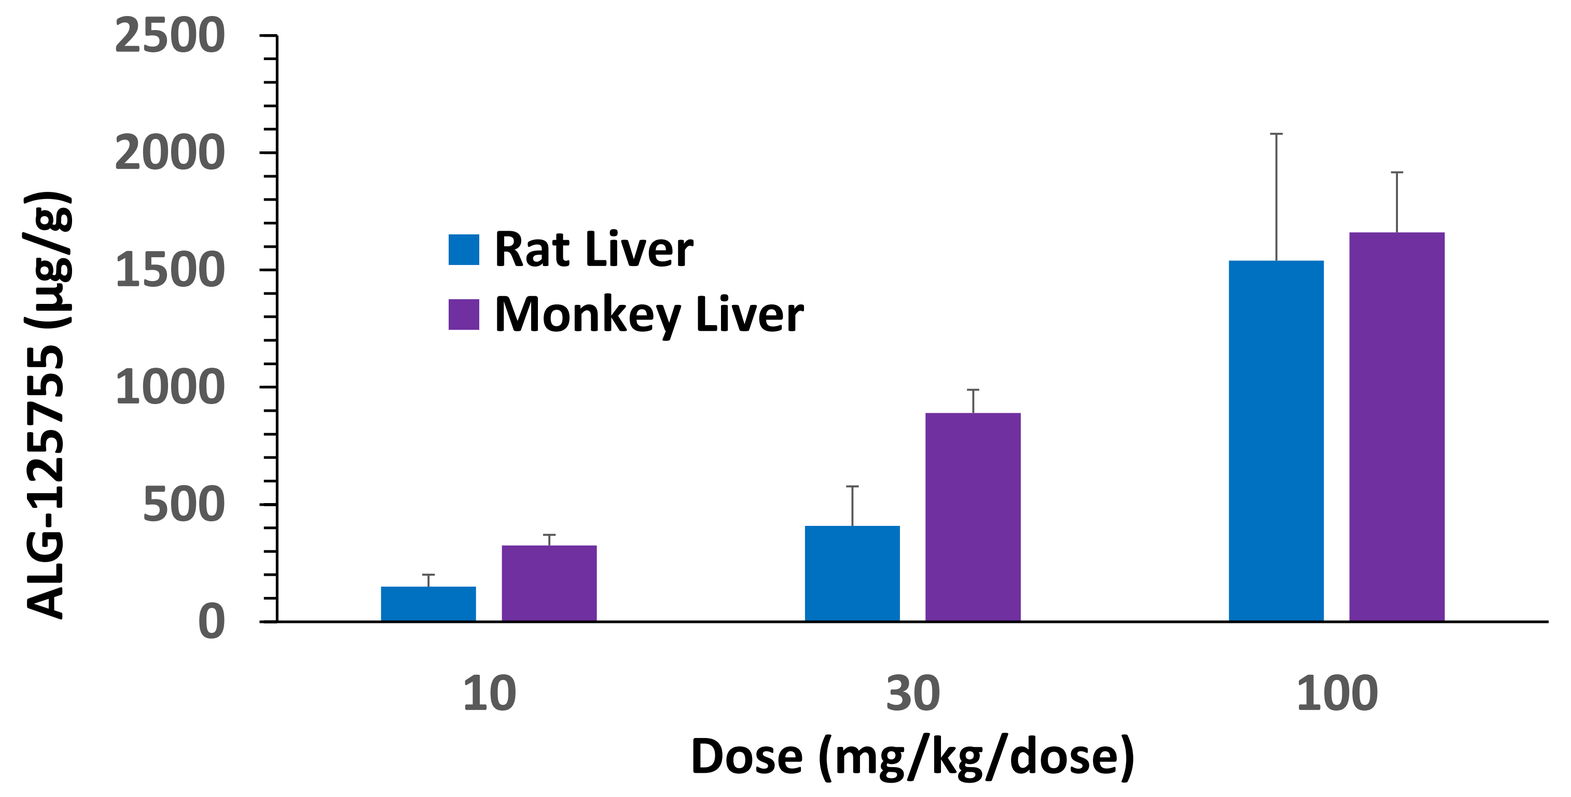
Rat Liver
Monkey Liver
ALG-125755 (µg/g)
Dose (mg/kg/dose)

Repeat SC Dose PK

- **Five weekly doses up to 100 mg/kg/dose were well tolerated in rat and monkey**
- **No sex difference in plasma exposures or tissue concentrations**
- **Nearly or greater than dose-proportional increase, and no accumulation of plasma exposures of ALG-125755**
- **Rapid, efficient and dose-dependent uptake in liver in both species**

Metabolism and Excretion

Metabolism

- **In all rat and monkey matrices the sense strand, either as GalNAc conjugate or as full sequence with loss of GalNAc moieties and linkers to varying degree, was present suggesting that the oligonucleotide remained as a duplex**
- **The major analyte in liver and kidney was ALG-125755 and the major metabolite was ALG-126144. Minor metabolites of the antisense strand formed by the loss of up to 9 nucleotides from the 3'-end and up to 8 nucleotides from the 5'-end were noted in both tissues**

Excretion

- **Urinary excretion within 168 hours postdose of intact ALG-125755 and ALG-126144 was low, accounting for 0.069% of the administered dose in monkeys following a single SC dose**

Pharmacokinetic/Pharmacodynamic Correlation in AAV-HBV Mouse Model

Figure 4. Correlation of liver concentrations at steady state to maximum HBsAg reduction

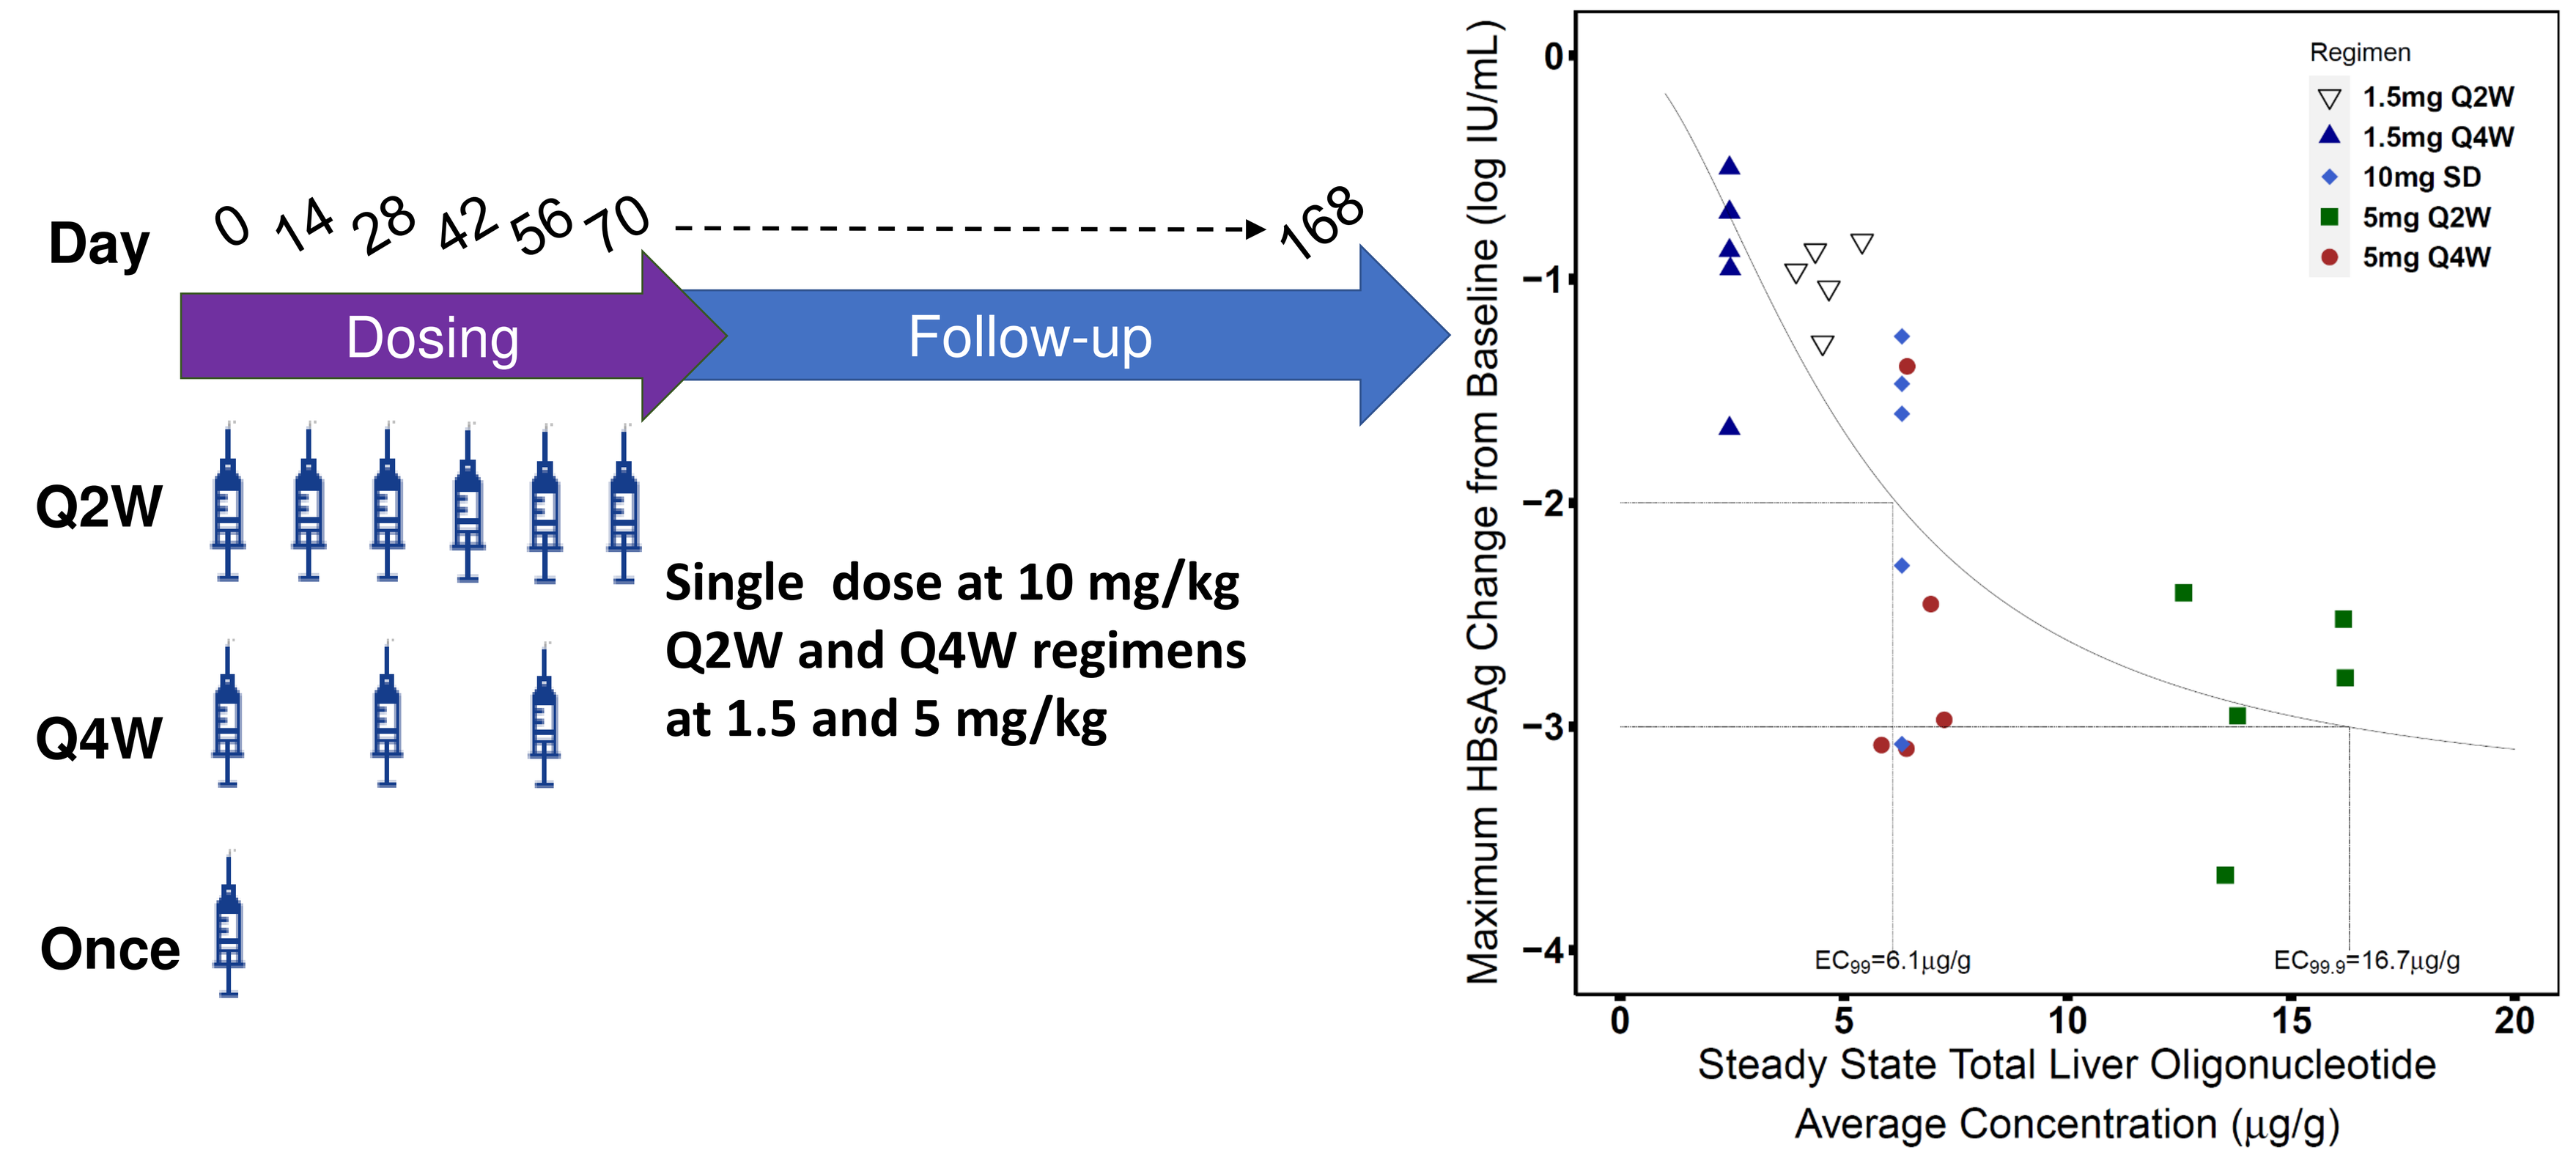
Day
0 14 28 42 56 70
168
Dosing
Follow-up
Q2W
Q4W
Once
Single dose at 10 mg/kg
Q2W and Q4W regimens
at 1.5 and 5 mg/kg
Regimen
1.5mg Q2W
1.5mg Q4W
10mg SD
5mg Q2W
5mg Q4W
Maximum HBsAg Change from Baseline (log IU/mL)
EC_{99}=6.1µg/g
$EC_{99.9}$=16.7µg/g
Steady State Total Liver Oligonucleotide
Average Concentration (µg/g)

- **A dose dependent inhibition of plasma HBsAg was observed with ALG-125755 in the model. Reductions in HBsAg levels were sustained for ≥70 days post-last dose (Fitzgerald et al Poster number SAT386, EASL 2022).**
- **Steady-state liver concentrations were projected using a population PK model based on sparse tissue concentrations of total siRNA for all dose groups**
- **Based on the analysis using a population PK and PK/PD I_{max} model, total liver oligonucleotides C_{avg} to achieve 99% (EC_{99}) and 99.9% ($EC_{99.9}$) HBsAg inhibition were projected to be 6.1 and 16.7 µg/g, respectively**

Conclusions

ALG-125755 demonstrated desirable preclinical pharmacology and PK/PD properties, including a long liver half-life that favors monthly or less frequent dosing in human. ALG-125755 is being advanced into the clinic for the functional cure of CHB with anticipated dosing in healthy volunteers in Q4 2022.

Financial disclosure: all authors are current or former employees of Aligos Therapeutics Inc.