Safety, Pharmacokinetics, and Antiviral Activity of the S-Antigen Transport Inhibiting Oligonucleotide Polymers (STOPS™) Drug Candidate ALG-010133 in Subjects with Chronic Hepatitis B

E. GANE[1], K. AGARWAL[2], M.F. YUEN[3], A. JUCOV[4], C. SCHWABE[5], K. LE[6], S. WANG[6], C. WESTLAND[6], K. STEEL[7], Q. ZHANG[6], V. GOHIL[6], F. LAI[6], V. MEENAKSHI[6], L. BEIGELMAN[6], L. BLATT[6], T.I. LIN[6], S. CHANDA[6], M. MCCLURE[6], and J. FRY[6]

[1]University of Auckland, New Zealand; [2]Institute of Liver Studies, Kings College Hospital, United Kingdom; [3]University of Hong Kong, Hong Kong; [4]ARENSIA Exploratory Medicine, Republican Clinical Hospital and Nicolae Testemitanu State University of Medicine and Pharmacy, Moldova; [5]New Zealand Clinical Research, New Zealand; [6]Aligos Therapeutics Inc, United States; [7]SARPO Consulting, Australia

INTRODUCTION

Worldwide, more than 296 million people are affected by Chronic Hepatitis B (CHB) and ~820,000 people per year die from cirrhosis and hepatocellular carcinoma (HCC) due to CHB.[1] Long-term treatment with current standard of care for CHB, nucleos(t)ide analogues (NA) or pegylated interferon, suppresses HBV replication and reduces liver injury in most patients, but rarely results in functional cure, the goal of CHB treatment.[2] Therefore, there is a significant medical need for novel approaches to enhance functional cure rates.

ALG-010133 is an S-antigen Transport Inhibiting Oligonucleotide Polymer (STOPS™) molecule which, in vitro, substantially reduces Hepatitis B surface antigen (HBsAg) production and release from infected hepatocytes with single digit nanomolar potency. ALG-010133 was being developed as a potential component of a finite duration combination regimen to achieve higher rates of functional cure. Study ALG-010133-101 (NCT04485663) was a 3-part, multicenter, double blind, randomized, placebo-controlled Phase 1 study designed to determine the preliminary safety, pharmacokinetics (PK) and antiviral activity of ALG-010133 in healthy volunteers (HVs) and CHB subjects. In Parts 1 and 2, single and multiple subcutaneous (SC) doses were generally well-tolerated in HVs.[3] Part 3 data evaluating the drug in CHB subjects are presented here.

AIM

To evaluate the safety, PK, and antiviral activity of multiple SC doses of ALG-010133 in CHB subjects.

METHODS

- Subjects in Part 3 of Study ALG-010133-101 received 12 weekly SC doses of ALG-010133 or placebo. Each cohort consisted of 10 CHB subjects randomized 4:1 to ALG-010133 or placebo. Key eligibility criteria for Part 3 of this study were:
 - Virologically suppressed (VS; HBV DNA <20 IU/mL) on stable NA treatment for ≥6 months
 - Hepatitis B e-antigen (HBeAg) negative
 - Alanine aminotransferase (ALT) and aspartate aminotransferase (AST) ≤1.2x upper limit of normal (ULN) and HBsAg ≥2 $\log_{10}$ IU/mL at screening
 - Metavir Score ≤F2
- Assessments of safety (adverse events [AEs], vital signs, electrocardiogram [ECG], physical examinations, and laboratories), viral kinetics (HBsAg, HBV DNA/RNA) and plasma/urine PK were systematically collected and analyzed.
- Blood samples were assessed for ALG-010133 plasma concentrations using validated hybridization-based Anion-Exchange High Performance Liquid Chromatography (AEX-HPLC) method coupled to a fluorescence detector and/or LC-MS/MS method.

RESULTS

DOSE LEVELS EVALUATED

- Cohort 1: 120 mg ALG-010133 or placebo
- Cohort 2: 200 mg ALG-010133 or placebo
- Cohort 3: 400 mg ALG-010133 or placebo

BASELINE CHARACTERISTICS

The baseline characteristics were generally similar across treatment groups and typical for a VS CHB population

	120 mg ALG-010133 N = 8	200 mg ALG-010133 N = 8	400 mg ALG-010133 N = 8	Placebo N = 7*
Age (years), mean (SD)	51.6 (10.6)	49.1 (8.9)	45.5 (8.4)	44.7 (12.8)
% Male	63%	50%	63%	71%
% White	88%	13%	38%	57%
BMI (kg/m^2), mean (SD)	26.9 (5.8)	24.4 (6.4)	28.5 (3.9)	23.5 (3.2)
HBsAg ($\log_{10}$ IU/mL), mean (SD)	3.6 (0.4)	3.6 (0.3)	3.5 (0.5)	3.1 (0.6)

SD = standard deviation *N of 7 due to over-enrollment of Cohort 3 by 1 subject as allowed per protocol

PHARMACOKINETICS

Plasma ALG-010133 exposures increased more than dose proportionally, with moderate variability and minimal accumulation.

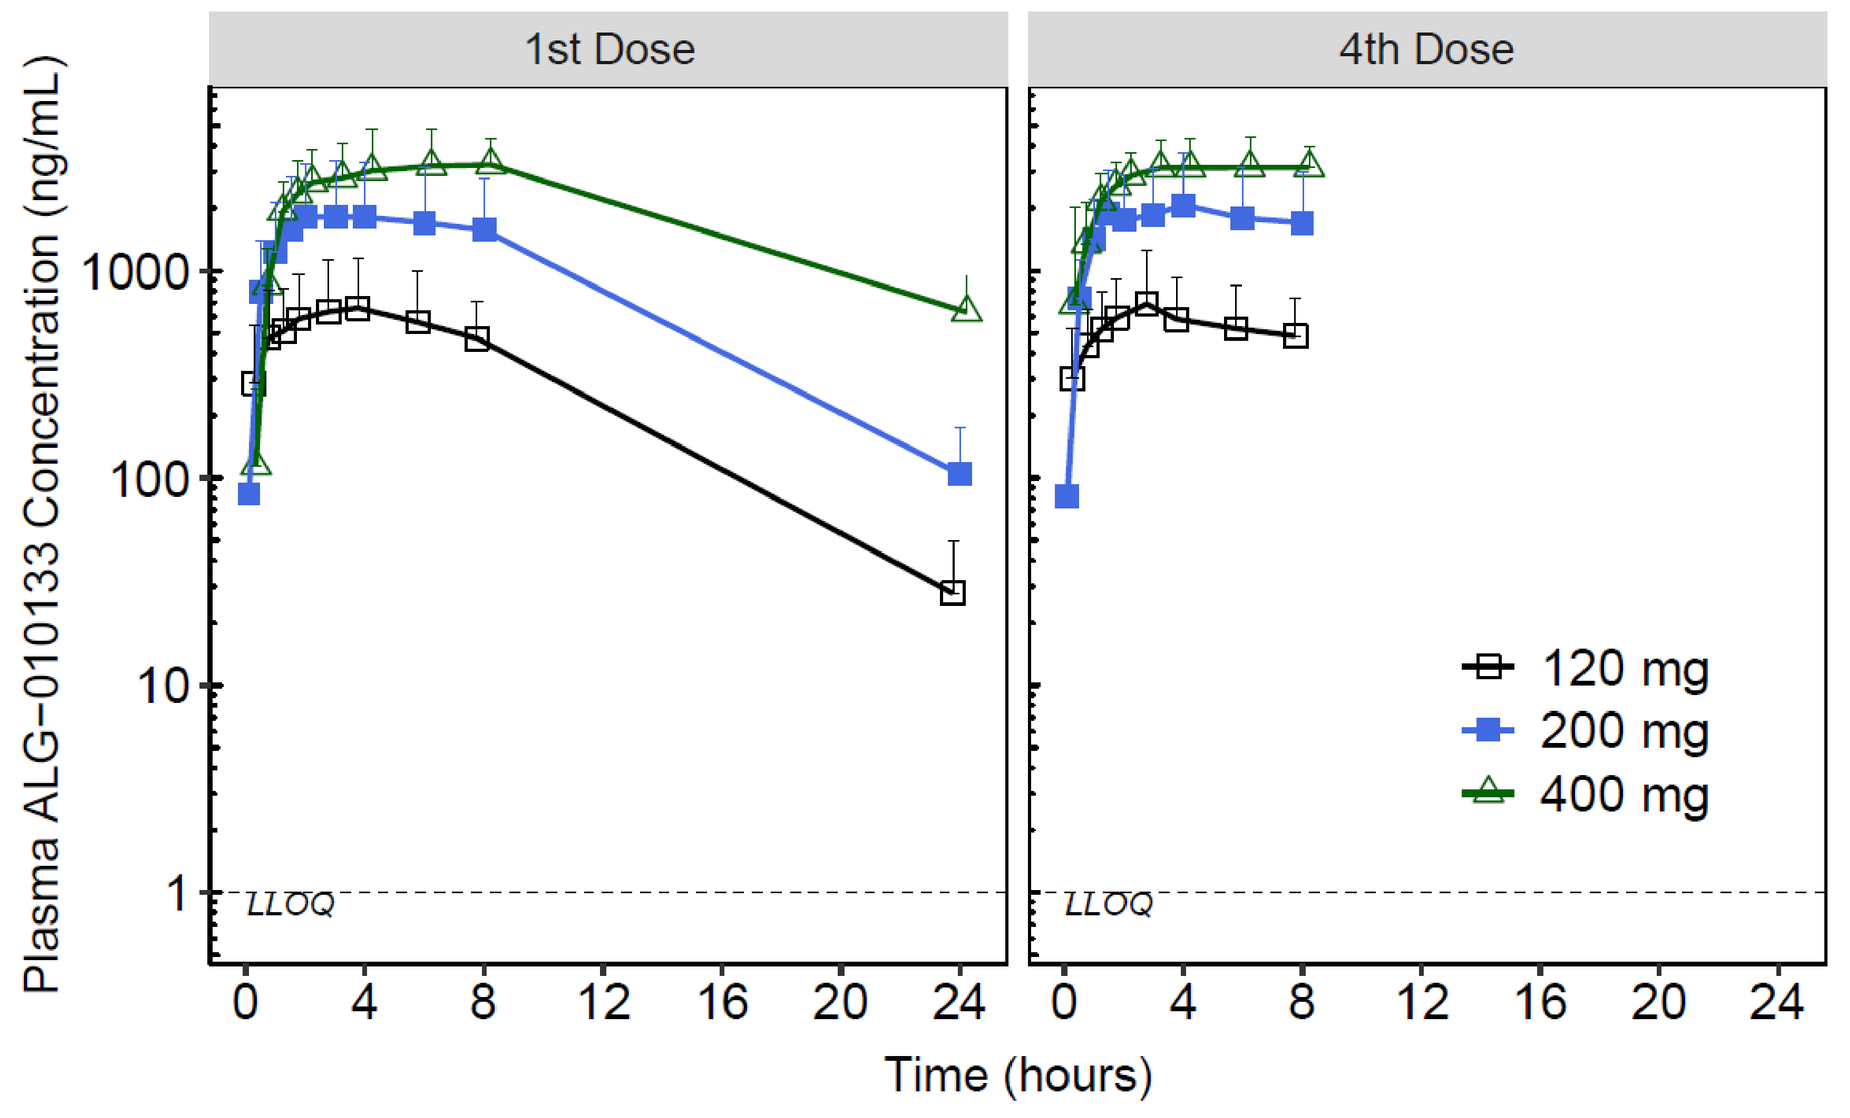

1st Dose
4th Dose
Plasma ALG-010133 Concentration (ng/mL)
Time (hours)
LLOQ
120 mg
200 mg
400 mg

Figure 1: Mean (SD) Plasma Concentration-Time Profiles of Multiple Doses of ALG-010133

Dose (mg)	Dose No.	BW (kg)	C_{max} (ng/mL)	t_{max} (hr)	AUC_{0-24} (ng.hr/mL)
120	1st Dose	78.9 (19.6)	593 (63.5)	4 (2,8)	5800 (51.5)
	4th Dose		611 (70.9)	3 (2,8)	7470 (57.6)
200	1st Dose	69.9 (31.2)	1520 (76.1)	2 (1.5,8)	15200 (68.6)
	4th Dose		1660 (71.8)	3 (1.5,6)	22800 (69.4)
400	1st Dose	80.2 (13.1)	3260 (44.9)	8 (4,8)	44300 (24.1)
	4th Dose		3390 (31.1)	4 (0.1,8)	57500 (38.5)

AUC_{0-24}=area under the plasma concentration versus time curve from 0 to 24 hours;
Data presented as geometric mean (Coefficient of Variation [CV]%), except t_{max}: median (minimum, maximum) and BW: mean (CV).

PREDICTED EFFICACIOUS DOSE

200 to 400 mg QW SC dosing regimens were projected to be efficacious by maintaining ~1-3x EC_{90} in liver at C_{trough}

SAFETY

Administration of 120 mg, 200 mg, and 400 mg of ALG-010133 once weekly (QW) for 12 weeks was generally well tolerated:

- One unrelated SAE of orchitis (medical history of epididymo-orchitis)
- One TEAE (COVID-19 infection) leading to premature study drug discontinuation considered unrelated to study drug
- No clinically significant dose response in incidence of TEAEs.
- All TEAEs were mild (Grade 1) or moderate (Grade 2), except 1 severe (Grade 3) TEAE of injection site erythema (severity based only on surface area criteria of ≥100 cm^2; required no treatment and resolved despite continued study drug dosing) and above TEAE of orchitis (Grade 3).
- No clinically concerning ECG, vital sign or physical examination findings

Event, n (%)	120 mg ALG-010133 N = 8	200 mg ALG-010133 N = 8	400 mg ALG-010133 N = 8	Placebo N = 7
Any TEAE	5 (62.5)	5 (62.5)	8 (100)	3 (42.9)
TEAEs Leading to Study Drug Discontinuation	0	0	1 (12.5)	0
Serious TEAEs	0	1 (12.5)	0	0
Any TEAE Grade ≥3	1 (12.5)	1 (12.5)	0	0
TEAEs in ≥3 subjects across all doses				
Injection site erythema	3 (37.5)	1 (12.5)	1 (12.5)	0
Injection site bruising	1 (12.5)	1 (12.5)	1 (12.5)	1 (14.3)
Increased ALT [a]	1 (12.5)	1 (12.5)	2 (25)	0
Increased AST [a]	1 (12.5)	0	2 (25)	0
Injection site pruritus	0	2 (25)	1 (12.5)	0

a. None associated with symptoms or evidence of synthetic dysfunction (i.e., bilirubin, INR, albumin abnormalities)

- Treatment emergent ALT elevations were commonly observed (n=16) among actively treated subjects and appeared to be dose responsive (Figure 2). However, all were Grade ≤2 (<5x ULN), and none were associated with symptoms, bilirubin/INR abnormalities or premature discontinuation of study drug.

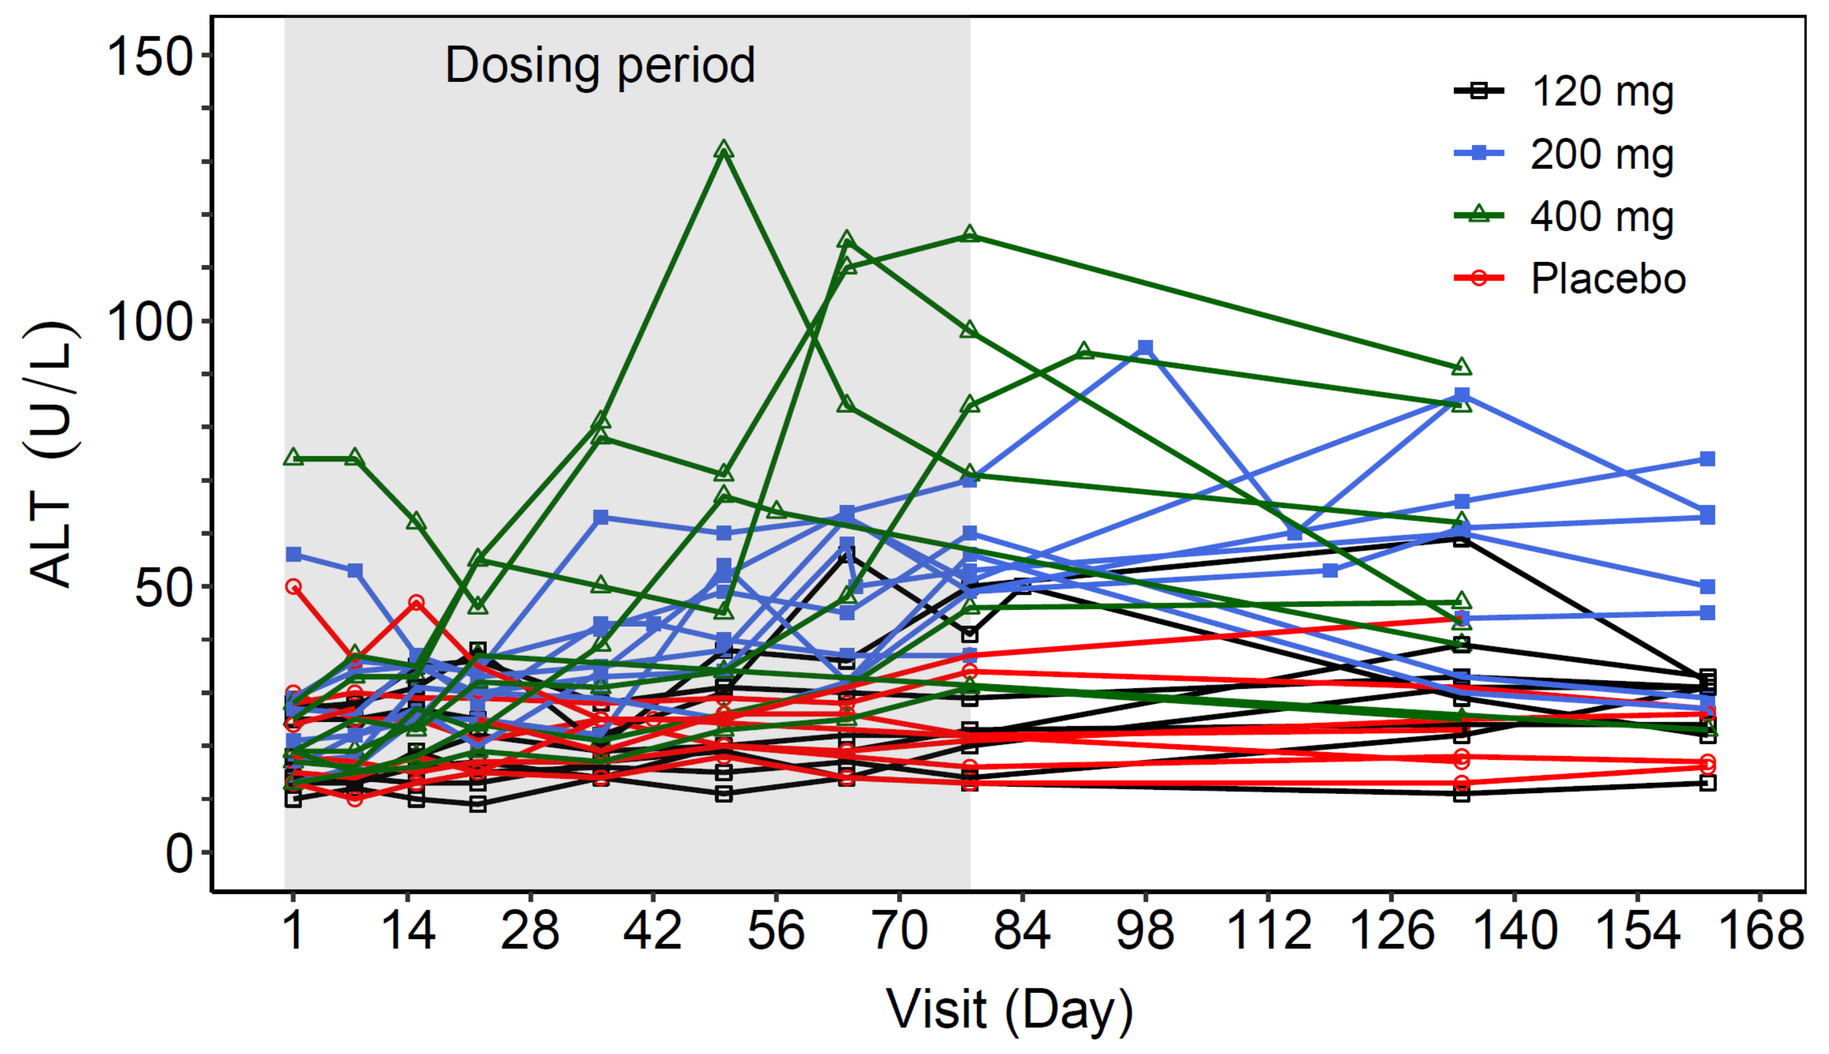

Dosing period
ALT (U/L)
Visit (Day)
120 mg
200 mg
400 mg
Placebo

Figure 2: ALT Levels Over Time from Baseline Through the End of Study

ANTIVIRAL ACTIVITY

Compared to baseline, the magnitude of HBsAg decline at Week 12 (Study Day 78) was <0.1 $\log_{10}$ IU/mL for placebo and across all ALG-010133 dose levels up to 400 mg

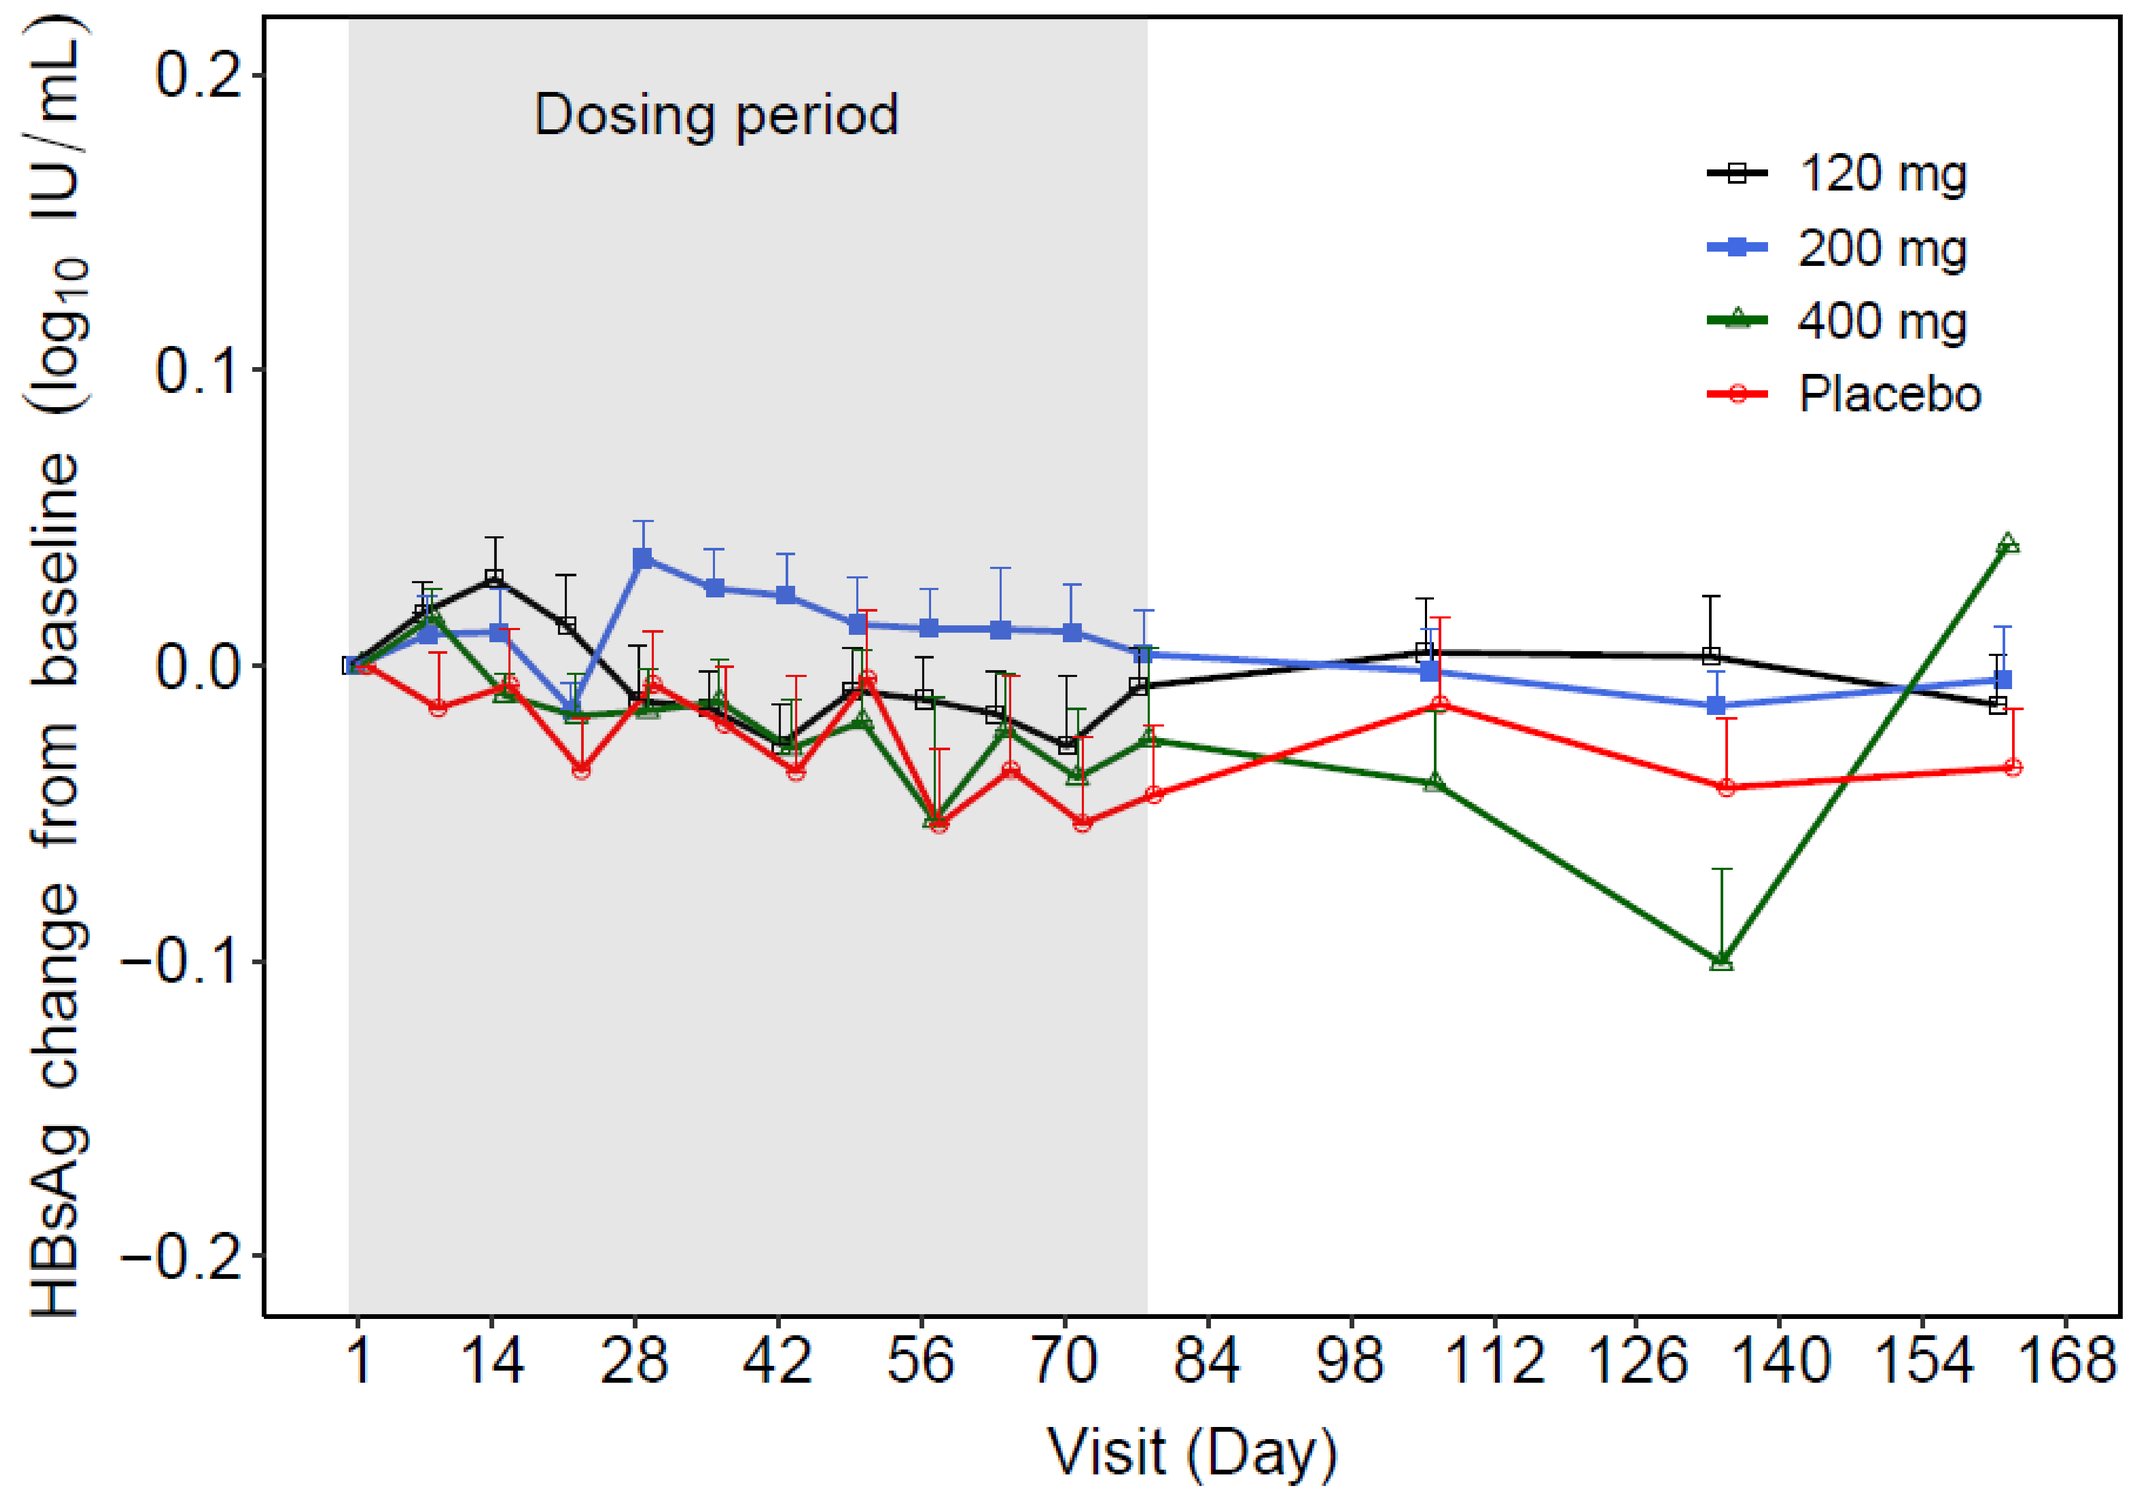

Dosing period
HBsAg change from baseline (log10 IU/mL)
Visit (Day)
120 mg
200 mg
400 mg
Placebo

Figure 3: Mean (SD) Serum HBsAg Level Change from Baseline Through the End of Study

CONCLUSIONS

- ALG-010133 was generally well tolerated with predictable PK properties when given to CHB subjects as 12 weekly SC doses of up to 400 mg
- Compared to placebo, no meaningful HBsAg reduction was observed with all dose levels of ALG-010133 up to 400 mg
- Further clinical development of ALG-010133 has been discontinued due to lack of efficacy
- The reasons for lack of efficacy are currently unclear. Unfortunately, the high *in vitro* potency exhibited by ALG-010133 and the prior clinical efficacy data for compounds of a similar class (i.e., nucleic acid polymers) did not translate in this study to robust antiviral effects in CHB patients treated with ALG-010133. Notably, a reliable *in vivo* model to test compounds of this class is not currently available, reflective of a mechanism of action that is not fully understood.

ACKNOWLEDGEMENTS

The authors wish to thank the subjects for participating in this clinical study. The Sponsor is grateful to the staff of the clinical sites, the Aligos study team, and to Novotech for assisting in the conduct of the study.

REFERENCES

1 WHO HBV fact sheet 2021.
2 Lok A et al. Hepatology Communications 2019;3(1):8-19.
3 Gane E et al. ILC 2021: Poster PO-1004.

CONTACT INFORMATION

Stanley Wang – swang@aligos.com