Incorporation of novel siRNA chemistries significantly improves the potency and durability of HBV siRNAs in the AAV-HBV mouse model

831 AASLD Nov. 12-15, 2021 The Liver Meeting® DIGITAL EXPERIENCE

Jin Hong, Saul Martinez Montero, Hua Tan, Vivek K. Rajwanshi, Hyunsoon Kang, Jacquelyn Sousa, Dawei Cai, Cheng Liu, Tilani De Costa, Vera Huang, Elen Rosler, Jerome Deval, Rajendra Pandey, Lawrence M. Blatt, Julian A. Symons, David B. Smith, Leonid N. Beigelman
Aligos Therapeutics, Inc., 1 Corporate Drive, 2nd Floor, South San Francisco, CA 94080

Contact: jhong@aligos.com Disclaimer: All authors are current or former Aligos Therapeutics, Inc., employees

BACKGROUNDS

The current standard of care for chronic hepatitis B (CHB) fails to reduce HBsAg needed to attain functional cure. HBV siRNA therapies have been shown to be effective in reducing HBsAg in CHB patients. For decades, 2'F and 2'OMe modified nucleotides have been the building blocks used to stabilize siRNA duplexes and maintain RNA-like properties. In this study, we applied multiple novel stabilization chemistries, including 5' phosphate mimetic end cap and unique 2'F mimic nucleotides to distinct HBV siRNAs. As a result, we achieved significant improvements in the extent and duration of HBsAg knockdown in the serum of the AAV-HBV mouse model.

The stability of HBV siRNAs were profiled in lysosome and snake venom phosphodiesterase (SVP) assays. Human Ago-2 assay was used to assess siRNA loading and cleavage of target RNA. Inhibition of HBsAg release was performed in the HepG2.2.15 cell line by transfection using RNAiMAX. The secreted HBsAg was quantified by ELISA. In the AAV-HBV mouse model, HBV siRNAs conjugated with proprietary GalNAc were administered subcutaneously (SC) with blood collections every 5 days for HBsAg and ALT assessments.

RESULTS

ALG-125755: Aligos HBV siRNA in development
ALG-125819: initial backup HBV siRNA

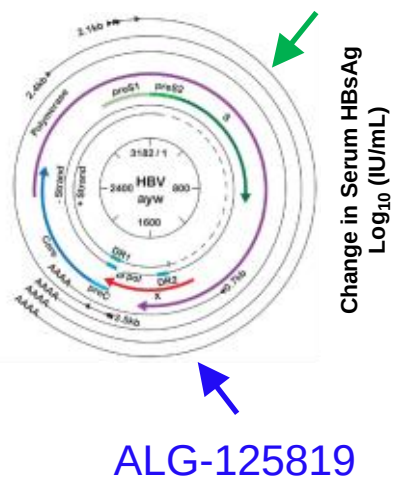
ALG-125755
HBV ayw
ALG-125819

Adapted from Lamontagne et. al. Hepatoma Res 2016;2:163-186

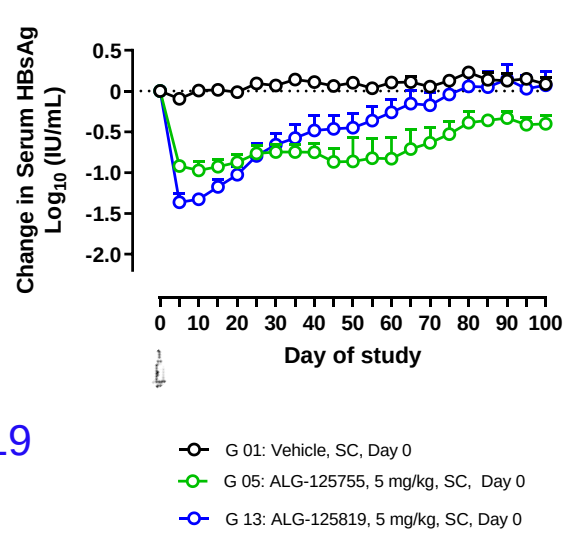
AAV-HBV Mouse Model: Serum HBsAg
Change in Serum HBsAg Log10 (IU/mL)
Day of study
G 01: Vehicle, SC, Day 0
G 05: ALG-125755, 5 mg/kg, SC, Day 0
G 13: ALG-125819, 5 mg/kg, SC, Day 0

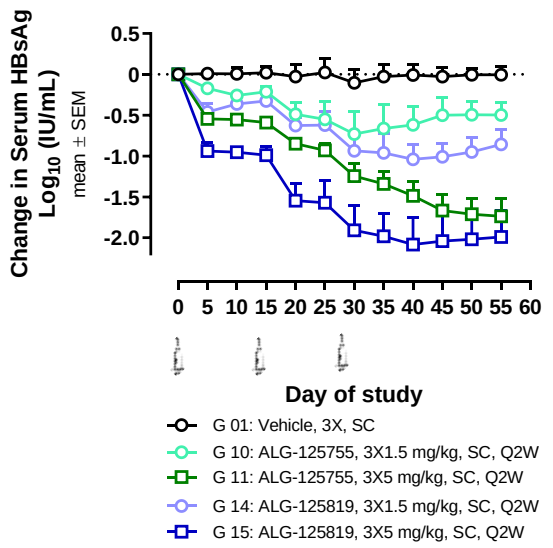
AAV-HBV Mouse Model: Serum HBsAg
Change in Serum HBsAg Log10 (IU/mL) mean ± SEM
Day of study
G 01: Vehicle, 3X, SC
G 10: ALG-125755, 3X1.5 mg/kg, SC, Q2W
G 11: ALG-125755, 3X5 mg/kg, SC, Q2W
G 14: ALG-125819, 3X1.5 mg/kg, SC, Q2W
G 15: ALG-125819, 3X5 mg/kg, SC, Q2W

Proprietary 2'F Mimic Modified ALG-125755: Modestly Improved Nuclease Stability and In Vivo Potency

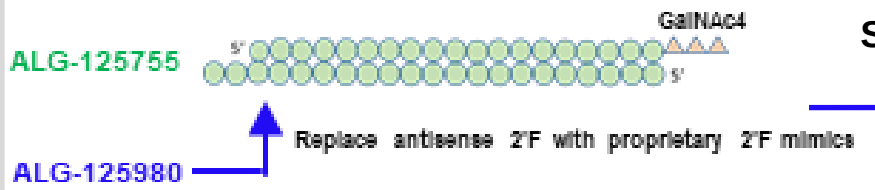
ALG-125755
GalNAc4
Replace antisense 2'F with proprietary 2'F mimics
ALG-125980

	HepG2.2.15 HBsAg Assay EC50	SVP Assay: % Remaining at 24 hr	Mouse Lysosomal Assay: % Remaining at 96 hr
Unconjugated ALG-125755	26 pM	0%	50%
Unconjugated ALG-125980	15 pM	50%	90%

Snake Venom Phosphodiesterase (SVP) Assay

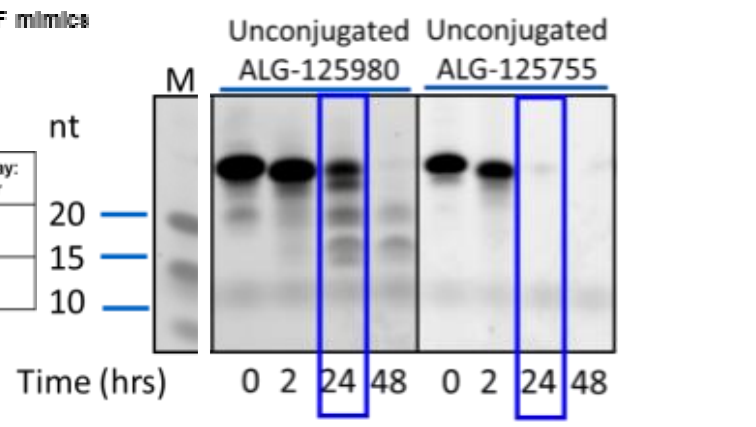
Unconjugated ALG-125980
Unconjugated ALG-125755
M
nt
20
15
10
Time (hrs) 0 2 24 48 0 2 24 48

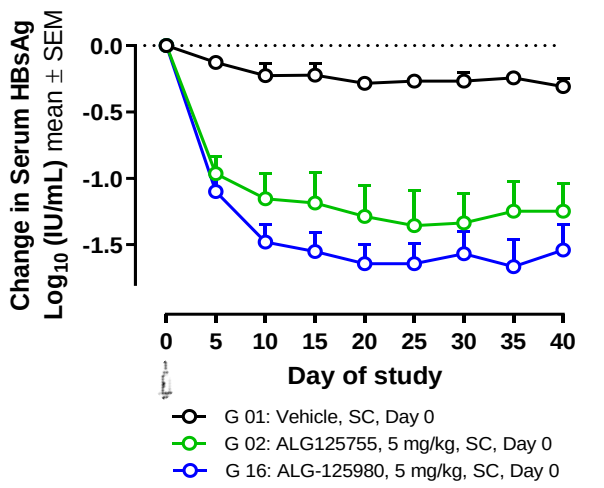
AAV-HBV Mouse Model: Serum HBsAg
Change in Serum HBsAg Log10 (IU/mL) mean ± SEM
Day of study
G 01: Vehicle, SC, Day 0
G 02: ALG125755, 5 mg/kg, SC, Day 0
G 16: ALG-125980, 5 mg/kg, SC, Day 0

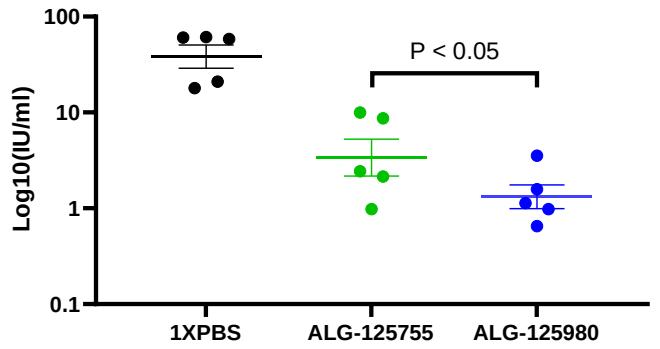
Liver HBsAg 40 Days Post Single 5 mg/kg Dose
Log10(IU/ml)
P < 0.05
1XPBS
ALG-125755
ALG-125980

Proprietary 5' End Cap Modified ALG-125819: Improved Ago-2 Activity

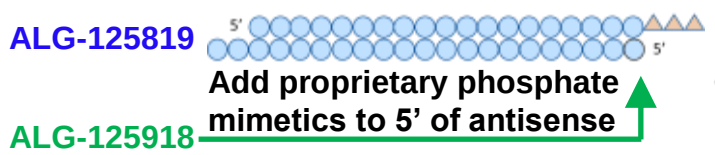
ALG-125819
GalNAc4
Add proprietary phosphate mimetics to 5' of antisense
ALG-125918

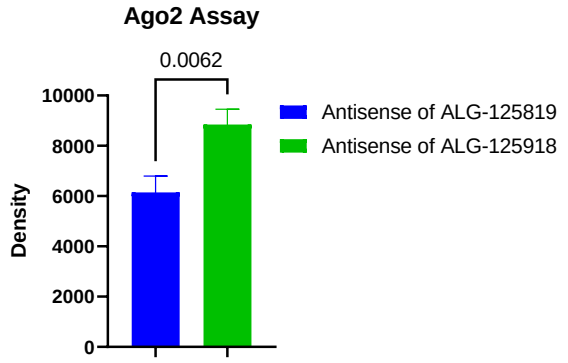
Ago2 Assay
0.0062
Density
Antisense of ALG-125819
Antisense of ALG-125918

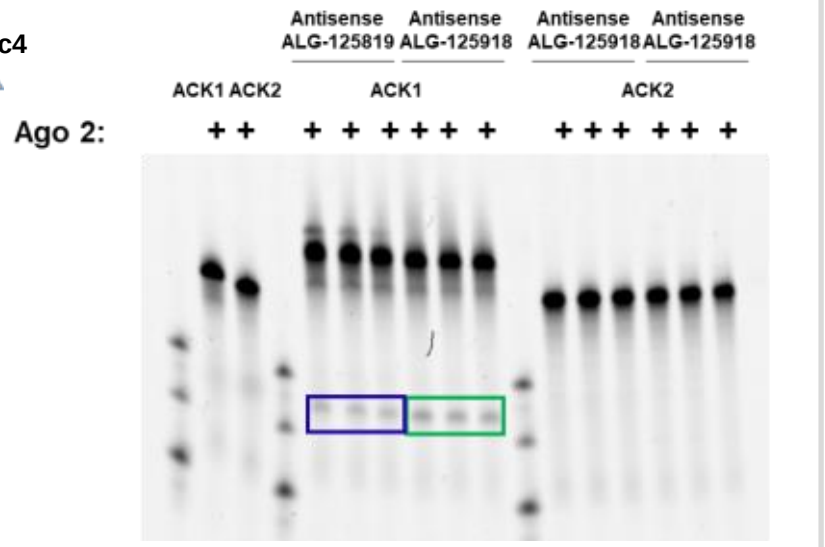
Antisense ALG-125819
Antisense ALG-125918
Antisense ALG-125918
Antisense ALG-125918
ACK1 ACK2
ACK1
ACK2
Ago 2:

ACK1: Cy5 HBV RNA; ACK2: Cy5 Non-HBV RNA

Results

ALG-125918 with Proprietary 5' End Cap Modification: Significantly Improved Maximum HBsAg Knockdown and Durability

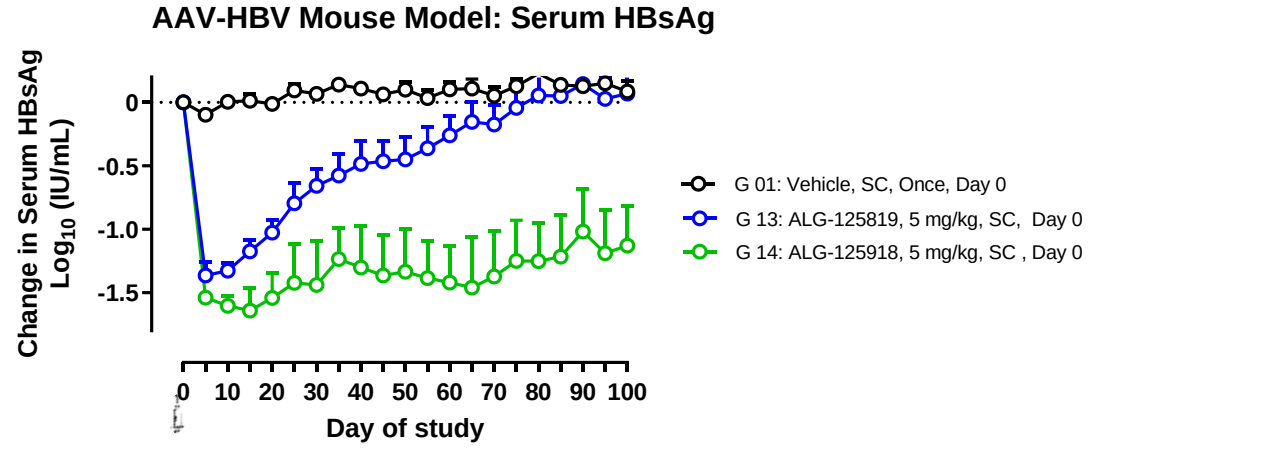
AAV-HBV Mouse Model: Serum HBsAg
Change in Serum HBsAg Log10 (IU/mL)
Day of study
G 01: Vehicle, SC, Once, Day 0
G 13: ALG-125819, 5 mg/kg, SC, Day 0
G 14: ALG-125918, 5 mg/kg, SC , Day 0

Further Improvement in ALG-125918 with 2'F mimics

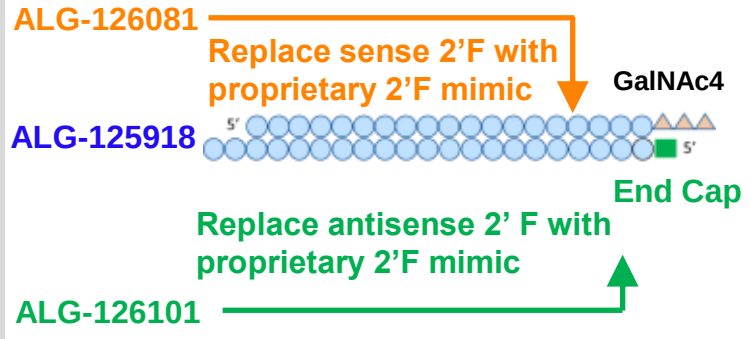
ALG-126081
Replace sense 2'F with proprietary 2'F mimic
GalNAc4
ALG-125918
End Cap
Replace antisense 2' F with proprietary 2'F mimic
ALG-126101

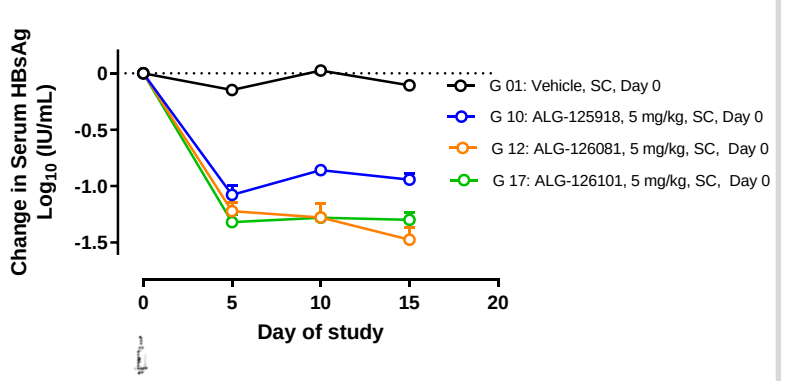
AAV-HBV Mouse Model: Serum HBsAg
Change in Serum HBsAg Log10 (IU/mL)
Day of study
G 01: Vehicle, SC, Day 0
G 10: ALG-125918, 5 mg/kg, SC, Day 0
G 12: ALG-126081, 5 mg/kg, SC, Day 0
G 17: ALG-126101, 5 mg/kg, SC, Day 0

1) **Aligos proprietary phosphate mimetic end cap improved siRNA loading and cleavage efficiency and significantly improved in vivo target knockdown depth and durability.**

2) **Aligos proprietary 2'F improved siRNA nuclease stability and in vivo potency.**

3) **Combination of end cap and 2'F mimics yielded very potent HBV siRNA backup molecules ALG-126081 and ALG-126101.**

AASLD

Slides are the property of the author and AASLD. Permission is required from both AASLD and the author for reuse.