Poster 1305

56th Annual International Liver Congress

Mechanism of Action of HBV S-antigen Transport-inhibiting Oligonucleotide Polymers (STOPSTM) Molecules

C. Cheng Kao, Yuchun Nie, Suping Ren, N. Tilani S. De Costa, Rajendra Pandey, Jin Hong, David B. Smith, Julian A. Symons, Leonid N. Beigelman, and Lawrence M. Blatt

Aligos Therapeutics, Inc., 1 Corporate Drive, South San Francisco, CA 94080

ALIGOS THERAPEUTICS

Abstract

Functional cure of chronic hepatitis B requires the elimination of serum hepatitis B virus surface antigen (HBsAg).[1] Nucleic acid polymers (NAPs), single-stranded phosphorothioated oligonucleotides, can reduce HBsAg in cultured cells and in patients.[2-4] STOPS, phosphorothioated oligonucleotides that contain novel chemistry, have enhanced inhibition of HBsAg by HBV-infected cells in vitro. This project aims to elucidate how STOPS cause the reduction of HBsAg.

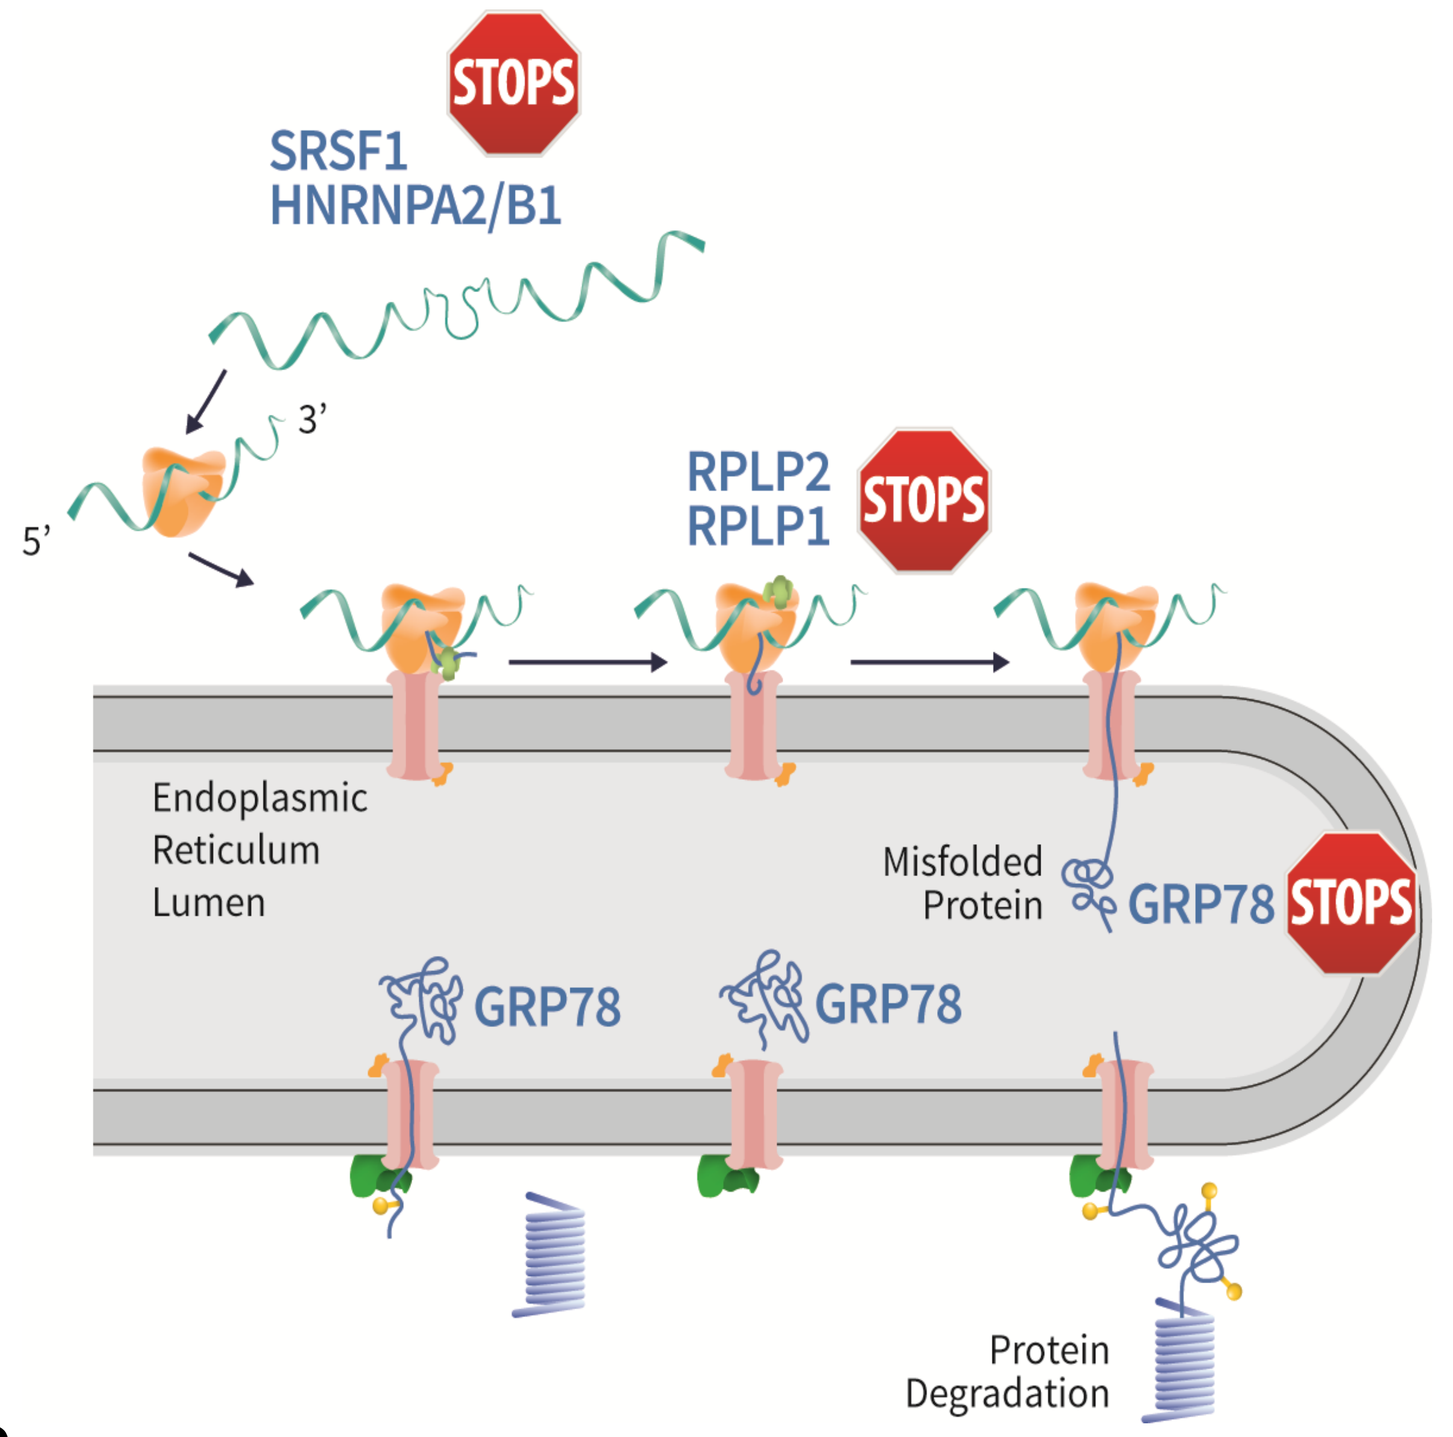

STOPS
SRSF1
HNRNPA2/B1
RPLP2
RPLP1
Endoplasmic Reticulum Lumen
Misfolded Protein
GRP78
Protein Degradation

The STOPS molecule ALG-10000 functions only when transfected into cells, where it localized to the cytoplasm. ALG-10000 reduced both intracellular and extracellular HBsAg. In addition, ALG-10000 reduced the level of HBV E-antigen, HBV polymerase, the core protein, total HBV RNA, and secreted rcDNA in HepG2.2.15 cells. ALG-10000 did not significantly co-localize with or bind to the HBsAg, indicating that STOPS acts through host-encoded factors. Five host proteins that function in RNA processing, translation, and protein folding/degradation were identified to bind ALG-10000: SRSF1, HNRNPA2B1, GRP78, RPLP1, and RPLP2. Notably, all of these factors have been previously described to function in viral replication and infection.[5-8] Silencing RNAs targeting these host factors reduced the levels of HBsAg and other HBV molecules known to be affected by STOPS. Interaction with multiple host factors is required to account for the inhibition of HBV infection. Finally, the introduction of ALG-10000, or the knockdown of GRP78, RPLP1, and RPLP2 in cells, increased the ubiquitination of HBsAg and proteasome-mediated degradation of HBsAg.

ALG-10000

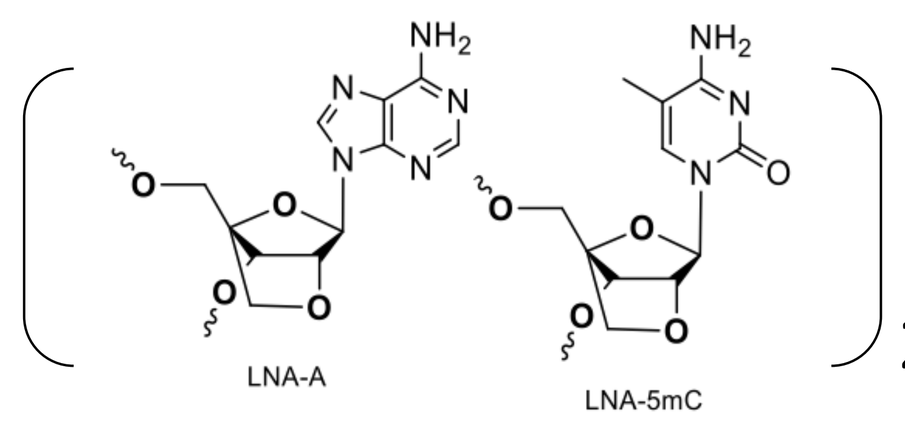

LNA-A
LNA-5mC
20

HBV-infected HepG2-NTCP cells

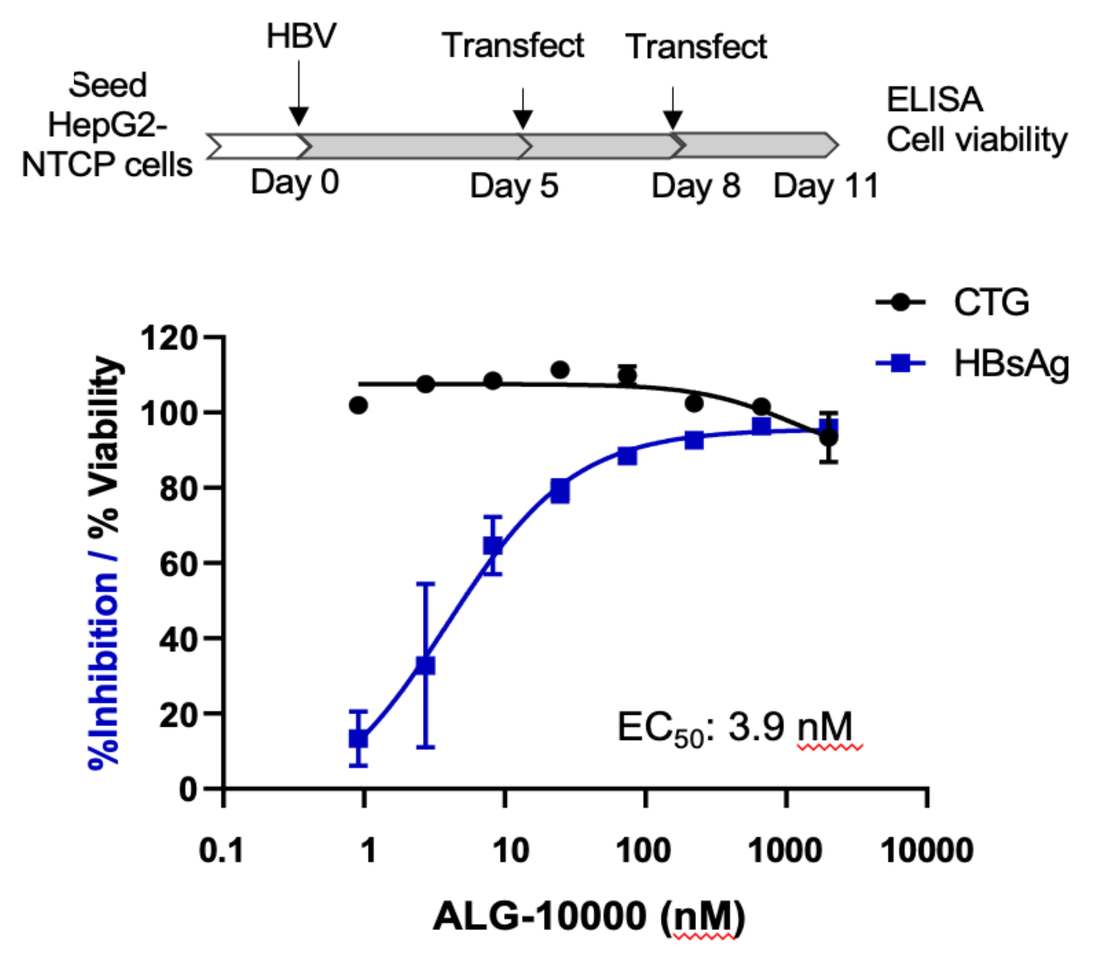

Seed HepG2-NTCP cells
HBV
Transfect
Transfect
ELISA
Cell viability
Day 0
Day 5
Day 8
Day 11
CTG
HBsAg
%Inhibition / % Viability
EC50: 3.9 nM
ALG-10000 (nM)

HepG2.2.15 cells that produce HBV

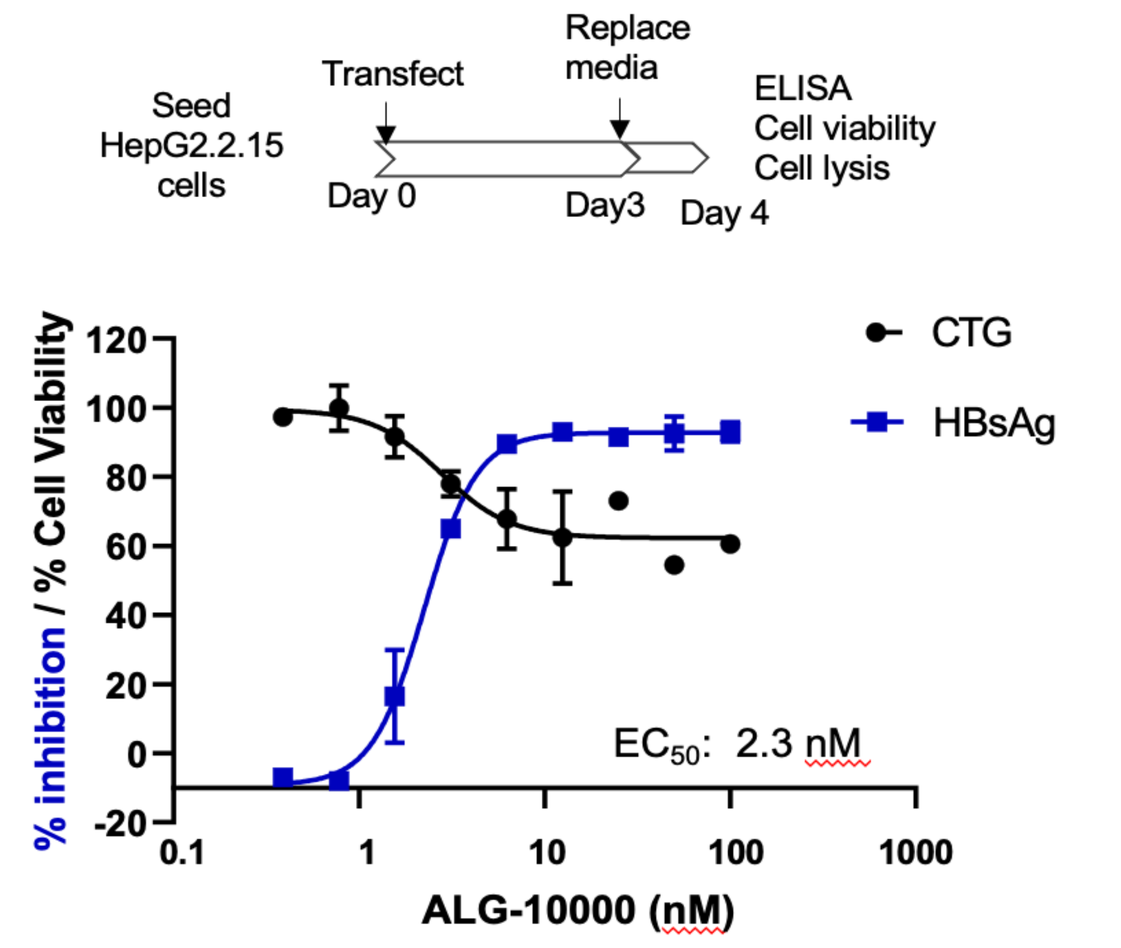

Seed HepG2.2.15 cells
Transfect
Replace media
ELISA
Cell viability
Cell lysis
Day 0
Day3
Day 4
CTG
HBsAg
% inhibition / % Cell Viability
EC50: 2.3 nM
ALG-10000 (nM)

Methods

STOPS and siRNAs were transfected into HepG2.2.15 cells that contain integrated HBV genomes using Lipofectamine RNAiMax. Proteins from HepG2.2.15 cell lysates that bind STOPS were identified using affinity chromatography, tandem mass spectrometry, and searches of the human proteome database. Fluorophore-labeled ALG-10000 in cells was visualized using a Leica SP6 microscope. HBsAg and other HBV proteins were quantified using ELISA and Western blotting. HBV nucleic acids were quantified using a Quantigene assay. Ubiquitination was detected by ELISA using a monoclonal antibody that was conjugated to horseradish peroxidase.

References and Disclosure

1. Yip TC et al. 2019. J. Hepatol. 70: 361.
2. Al-Mahtab M. *et al*. PLoS ONE (2016) , e0156667.
3. Bazinet M. *et al*. Fri_210 EASL (2019) Vienna.
4. Vaillant A. Antiviral Research 133 (2016) 32-40.
5. Paz S, Krainer AR, Caputi M. 2014. NAR 42: 13812.
6. Campos RK *et al*. 2020. NAR 48: 9872.
7. Guerens T, Bouhy D, Timmerman V. 2016. Hum. Genet. 135: 851.
8. Shu W. et al., 2020. Mol. Cell Biol. 40: e00475.

Disclosure: All authors are employees or consultants of Aligos Therapeutics, Inc.

ALG-10000 inhibits multiple HBV molecules but to different extents

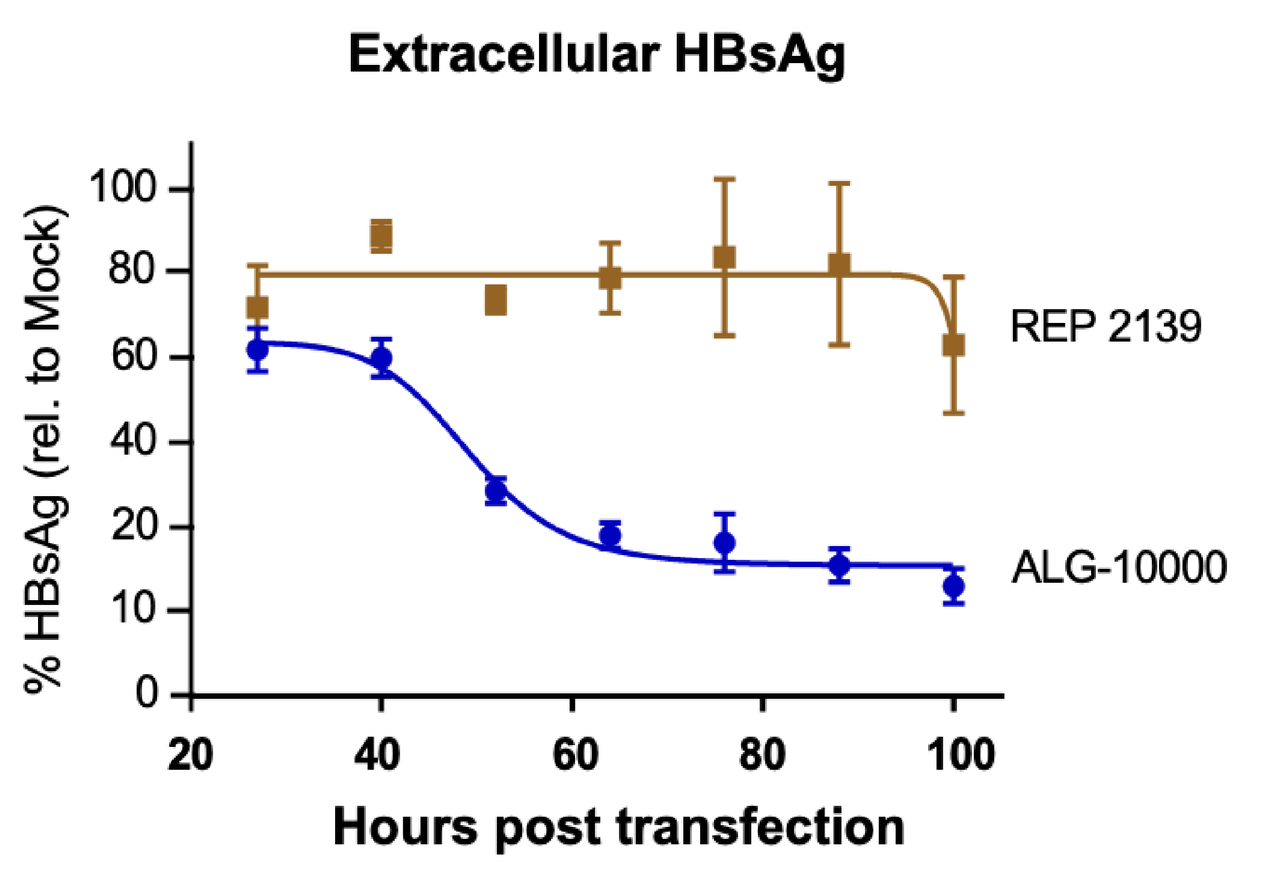

Extracellular HBsAg
% HBsAg (rel. to Mock)
REP 2139
ALG-10000
Hours post transfection

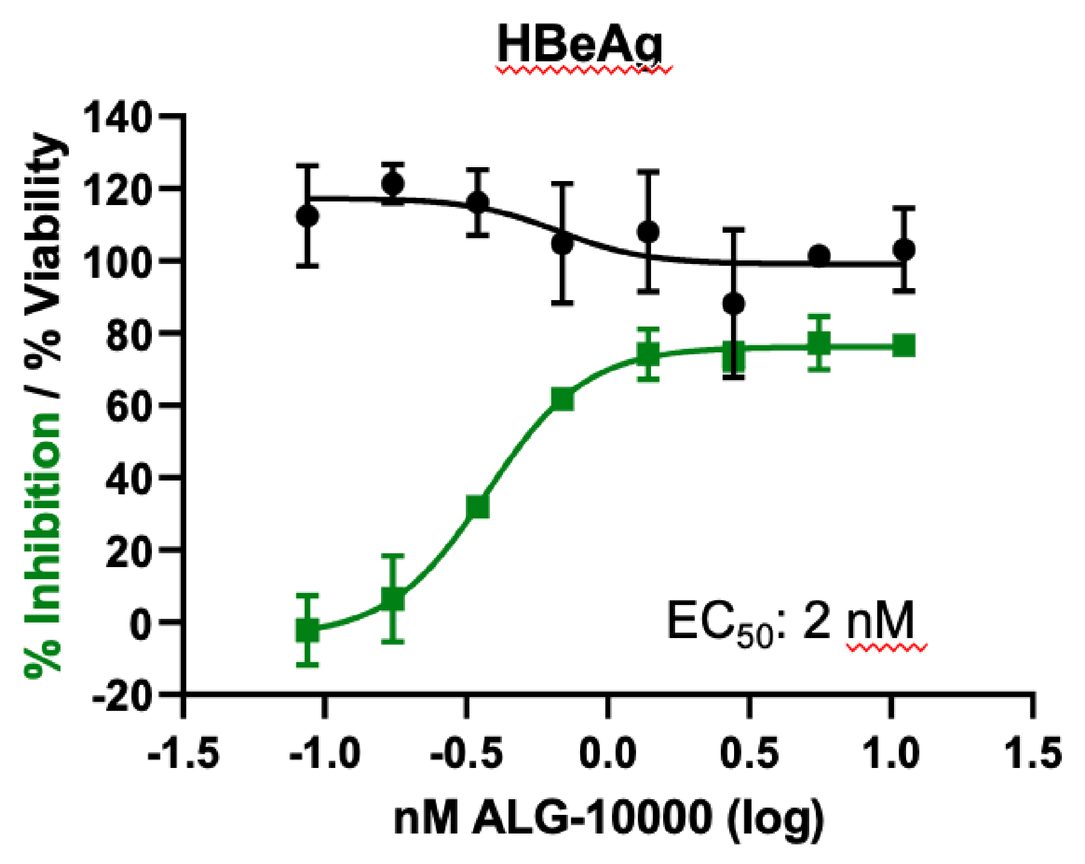

HBeAg
% Inhibition / % Viability
EC50: 2 nM
nM ALG-10000 (log)

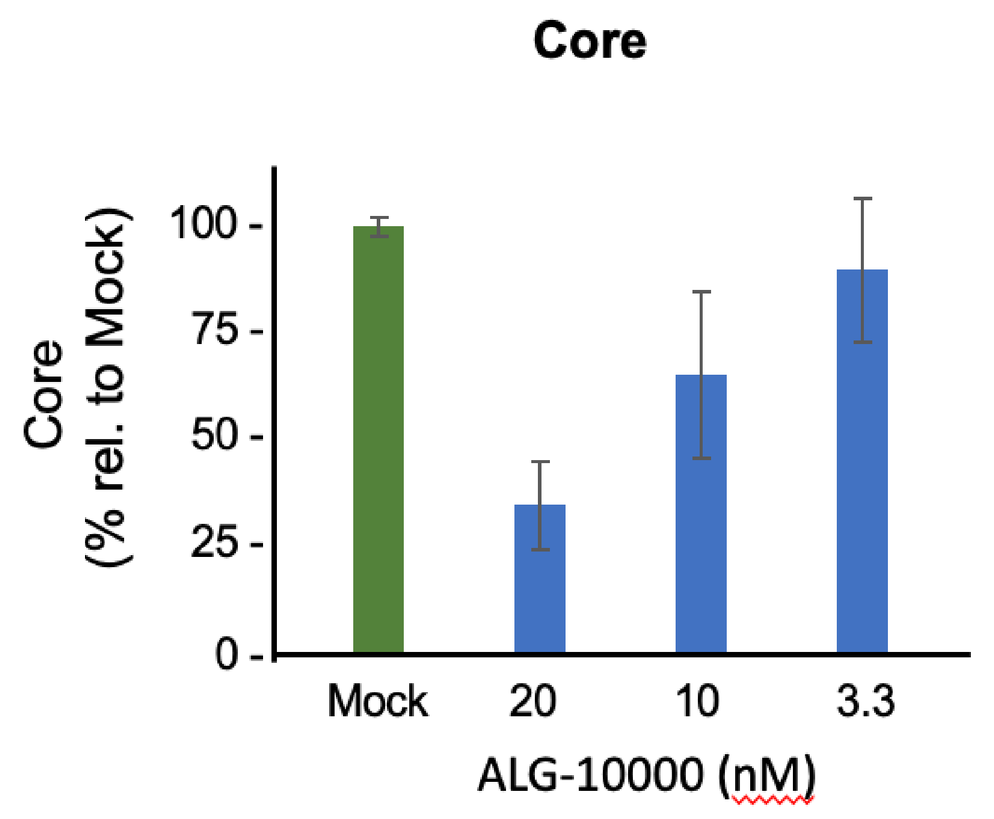

Core
Core (% rel. to Mock)
Mock
20
10
3.3
ALG-10000 (nM)

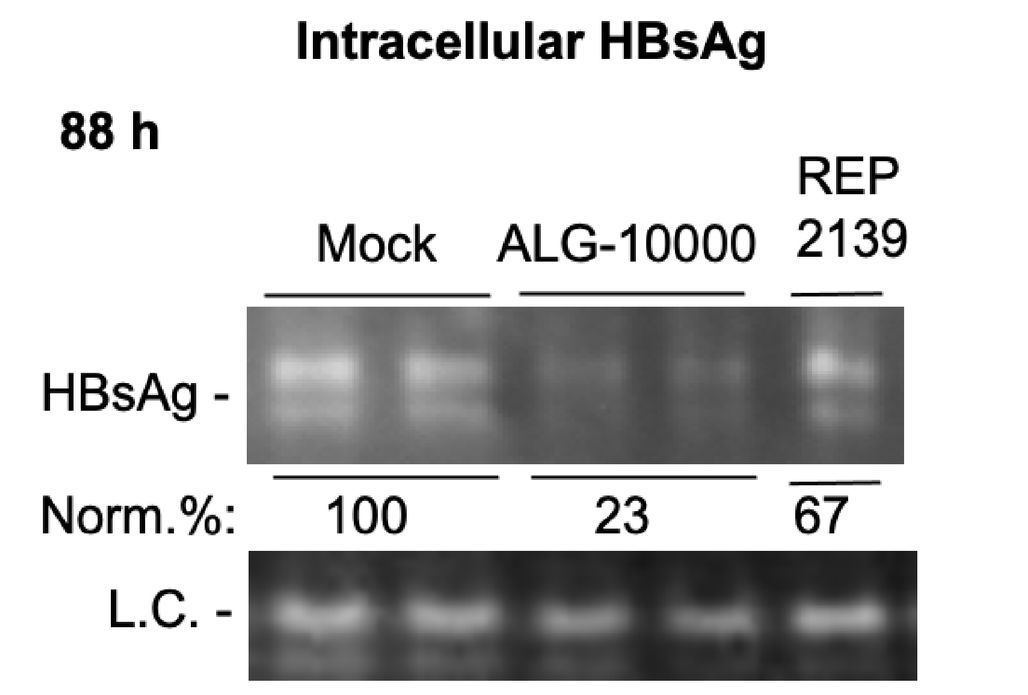

Intracellular HBsAg
88 h
Mock
ALG-10000
REP 2139
HBsAg -
Norm.%:
100
23
67
L.C. -

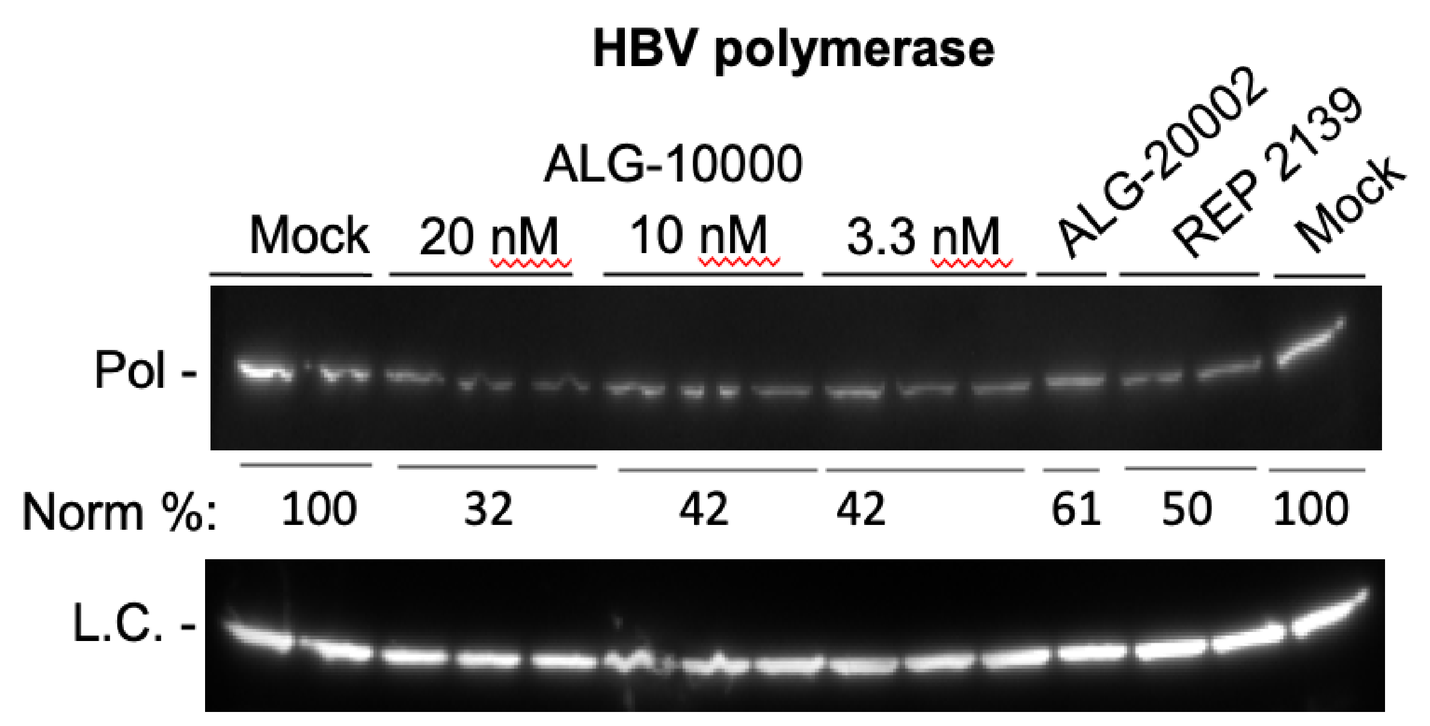

HBV polymerase
ALG-10000
Mock
20 nM
10 nM
3.3 nM
ALG-20002
REP 2139
Mock
Pol -
Norm %:
100
32
42
42
61
50
100
L.C. -

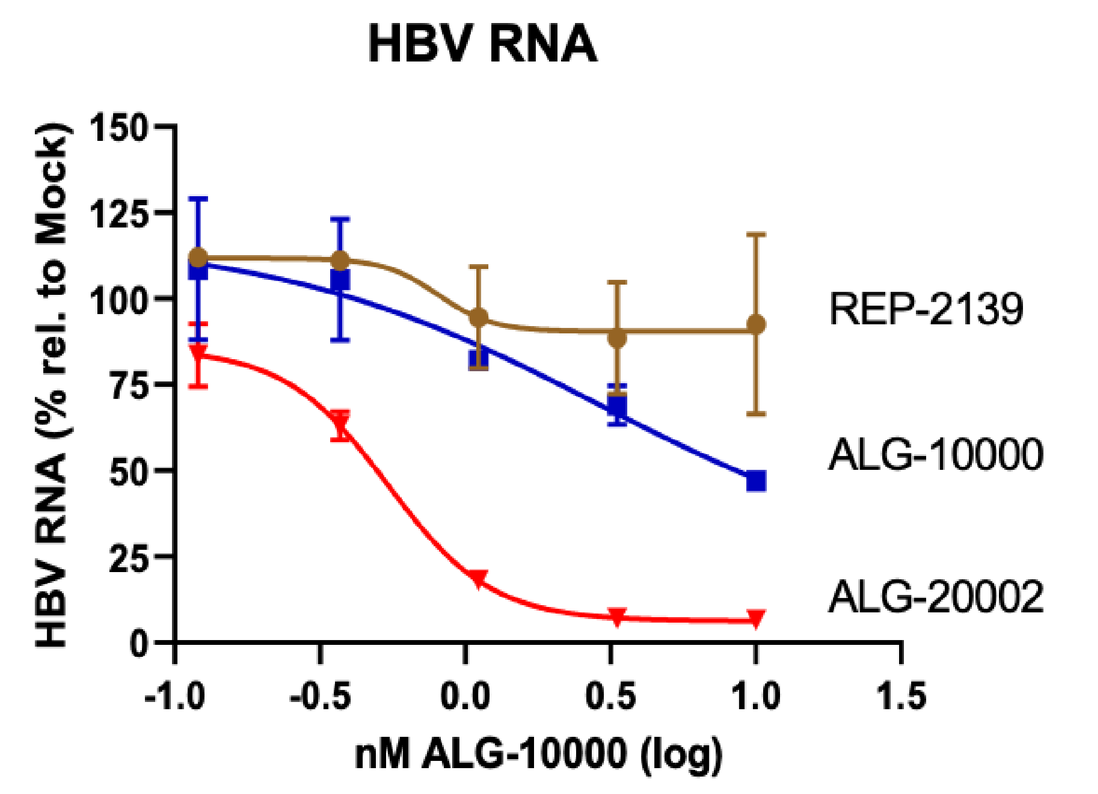

HBV RNA
HBV RNA (% rel. to Mock)
REP-2139
ALG-10000
ALG-20002
nM ALG-10000 (log)

STOPS have a complex mechanism of inhibition.

Inhibition of intracellular HBsAg suggests that inhibition precedes HBsAg trafficking and secretion.

ALG-10000 does not co-localize with HBsAg or activate nucleic acid-sensing immune receptors

Confocal microscopy for HBsAg and ALG-10000 localization

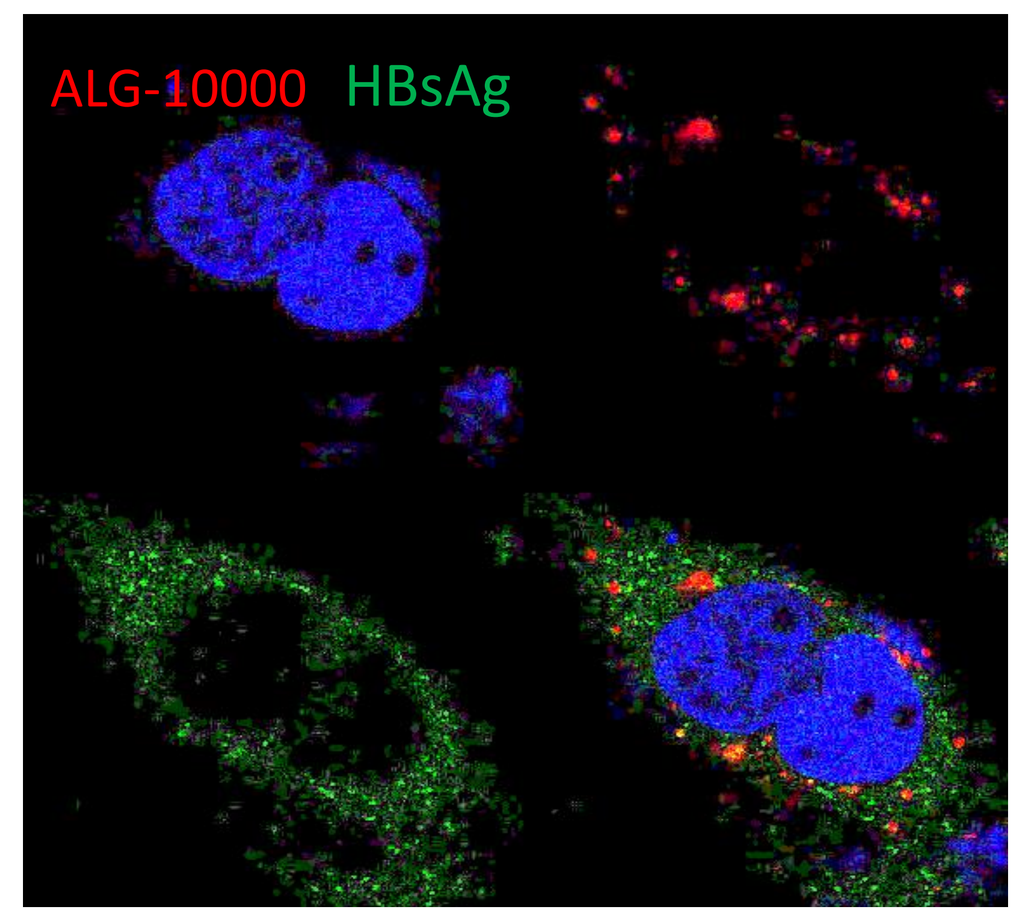

ALG-10000
HBsAg

Innate immune receptor reporter assays

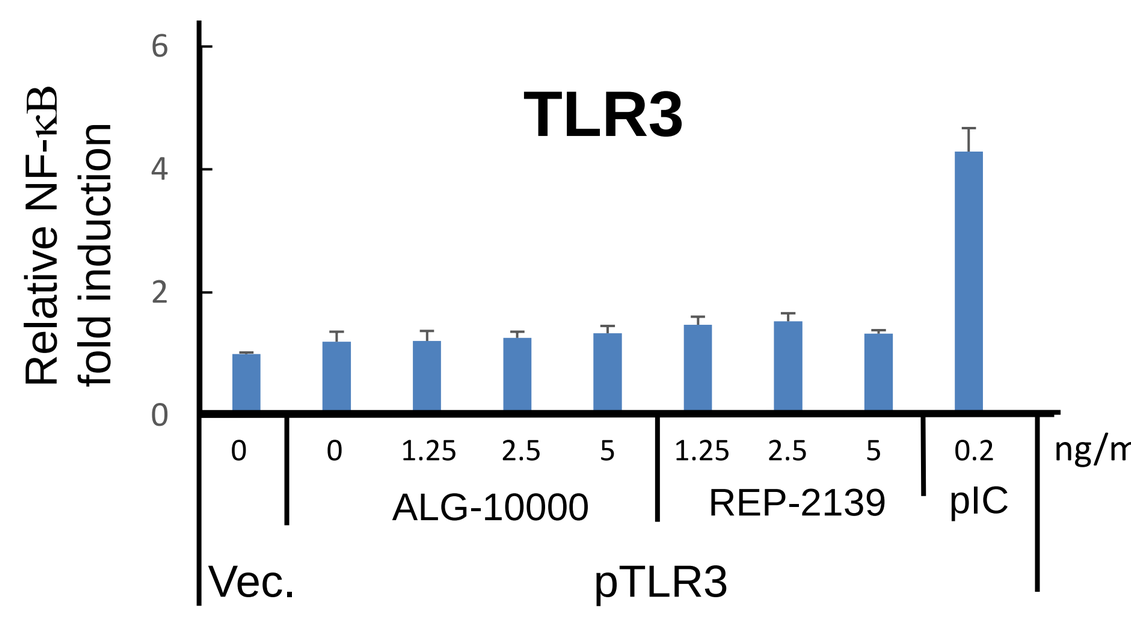

TLR3
Relative NF-κB fold induction
ALG-10000
REP-2139
pIC
ng/mL
Vec.
pTLR3

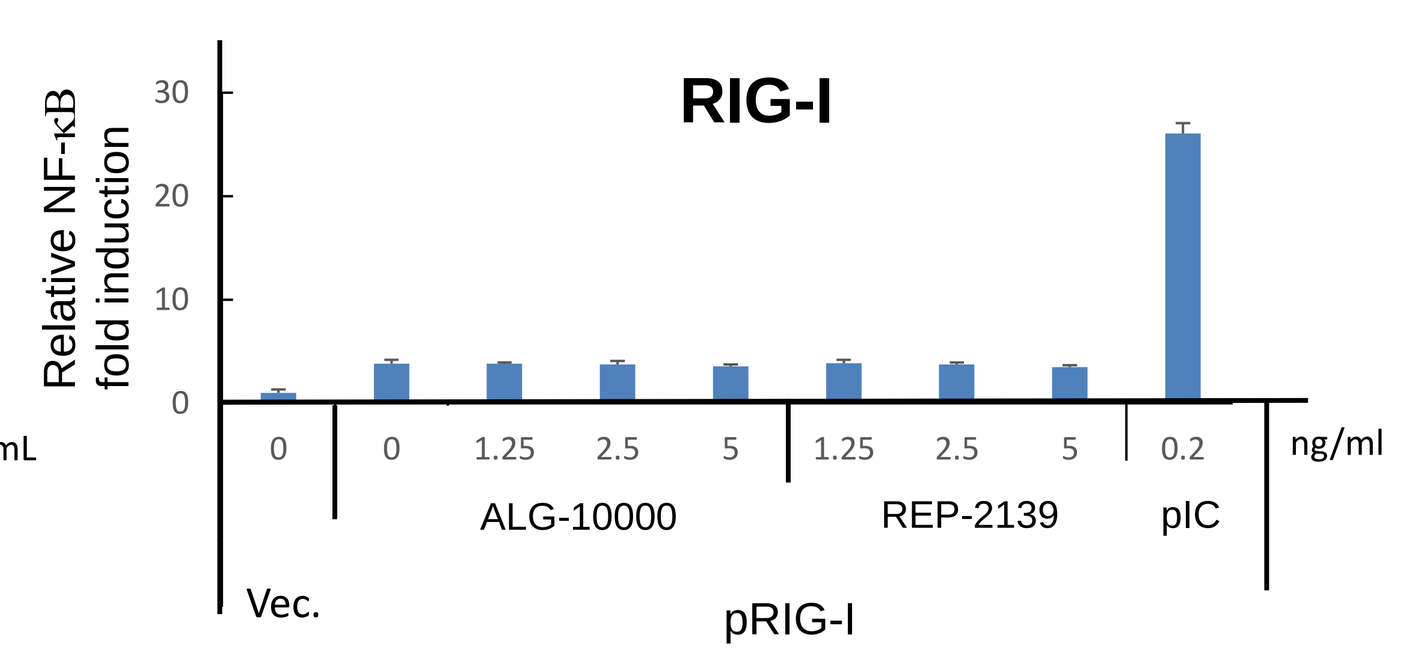

RIG-I
Relative NF-κB fold induction
ALG-10000
REP-2139
pIC
ng/ml
Vec.
pRIG-I

STOPS likely act through host factors.

ALG-10000 binds cellular proteins that function in gene expression

Top 21 proteins that bind the ALG-10000 resin; tandem MS/MS and bioinformatic analysis

Protein	Gene Name	# peptides	Confidence (%)
Nucleophosmin	NPM1	7	100.0
Histone H4	HIST1H4A	6	100.0
Endoplasmic reticulum chaperone BiP	GRP78/BiP/HSPA5	5	95.0
Histone H2B	HIST1H2BK	5	100.0
Histone H2AX	H2AFX	4	100.0
Nuclease-sensitive element-binding protein	1YBX1	4	100.0
Nucleolin	NCL	4	100.0
Peroxiredoxin 1	PRDX1	4	94.7
Protein-lysine 6 oxidase	LOX	3	100.0
Dermicidin	DCD	3	100.0
Small nuclear ribonucleoprotein Sm D1	SNRPD1	2	100.0
A-kinase anchor protein 11	AKAP11	2	95.2
Histone H3	H3F3A	2	95.0
Heterogeneous nuclear riboprotein A2B1	HNRNPA2B1	1	100.0
Calmodulin 2	CALM2	1	100.0
Serine/arginine-rich splicing factor 1	SRSF1	1	100.0
Protein SETSIP	SETSIP	1	100.0
Actin, cytoplasmic 1	ACTB	1	100.0
Prelamin A/C	LMNA	1	100.0
Thioredoxin	TXN	1	100.0
60S acidic ribosomal protein P2	RPLP2	1	100.0
ATP synthase-coupling factor 6, mitochondrial	ATP5PF	1	100.0

Several recombinant HF proteins can bind ALG-10000

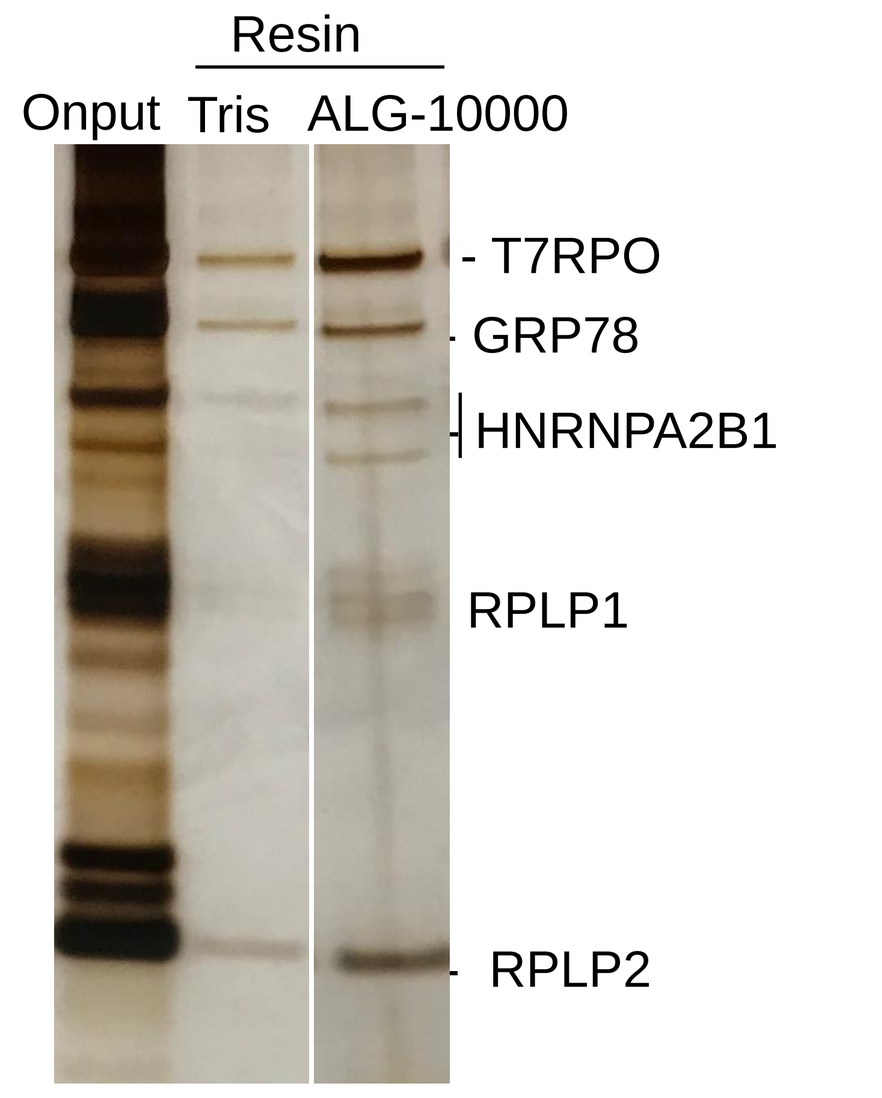

Resin
Onput
Tris
ALG-10000
T7RPO
GRP78
HNRNPA2B1
RPLP1
RPLP2

ALG-10000 reduces the levels of bound cellular proteins

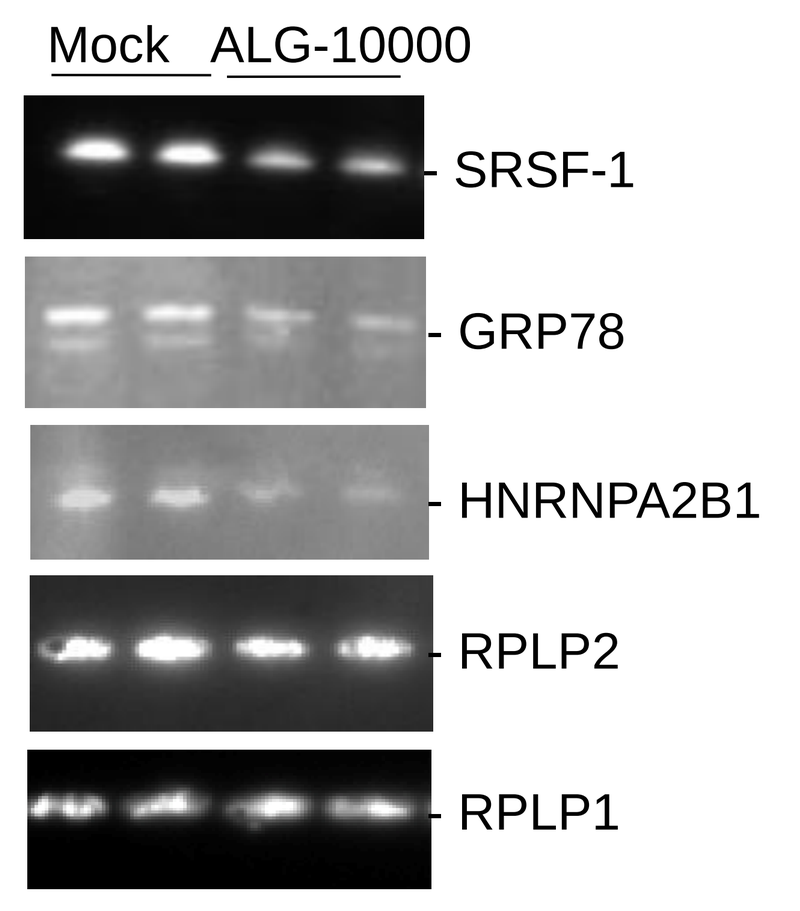

Mock
ALG-10000
SRSF-1
GRP78
HNRNPA2B1
RPLP2
RPLP1

Five of the cellular proteins that bind ALG-10000 are required for optimal HBsAg production. All 5 proteins have previously been reported to function in viral infections[5-8].

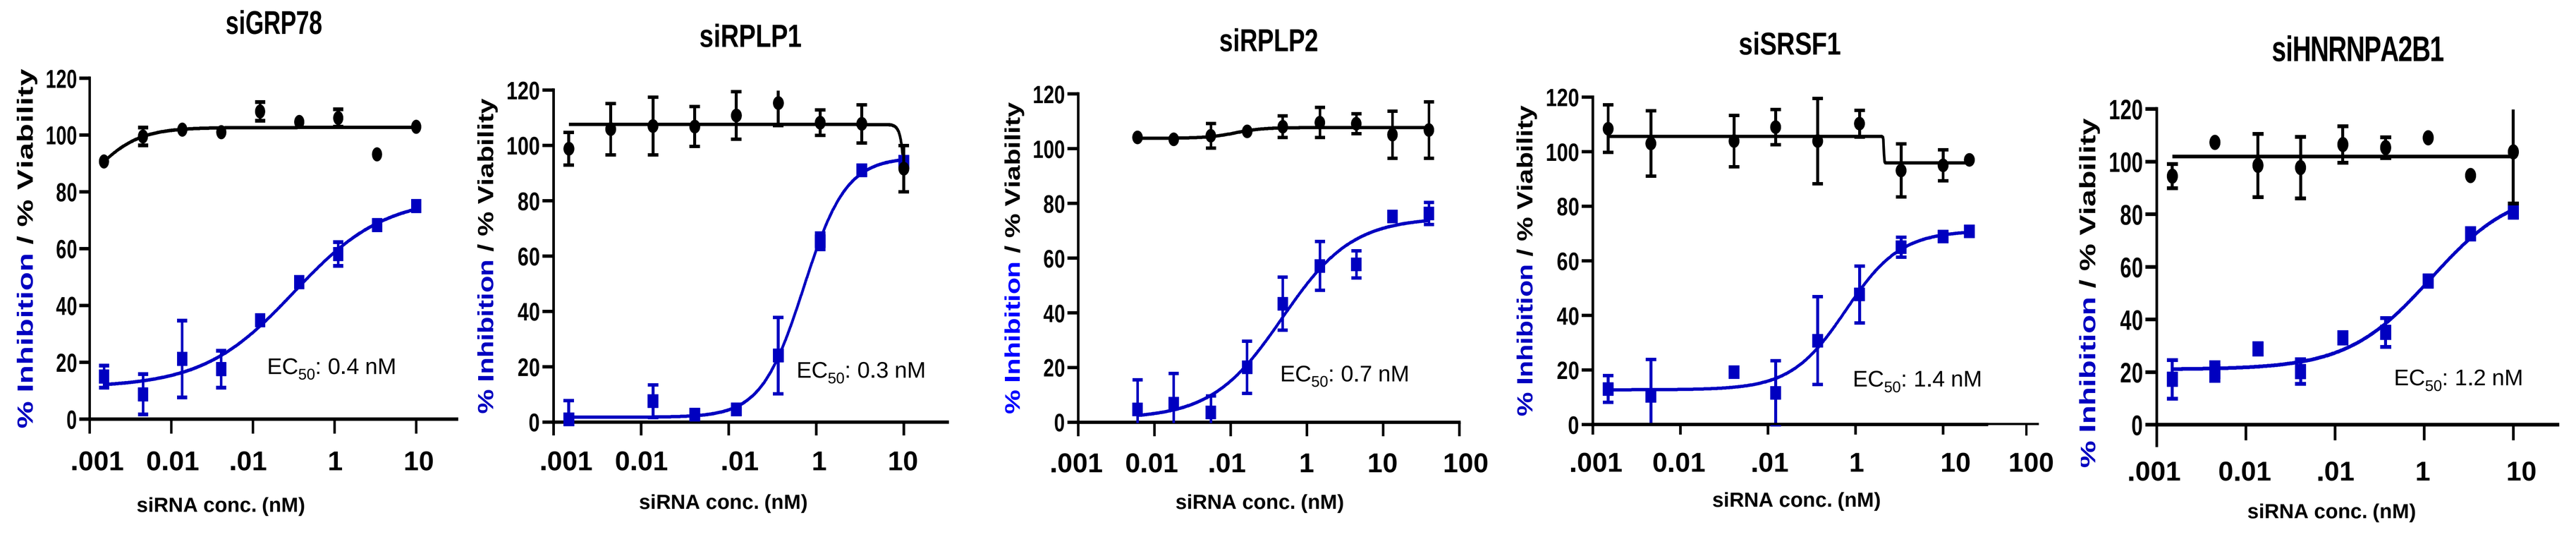

siGRP78
EC50: 0.4 nM
siRPLP1
EC50: 0.3 nM
siRPLP2
EC50: 0.7 nM
siSRSF1
EC50: 1.4 nM
siHNRNPA2B1
EC50: 1.2 nM
% Inhibition / % Viability
siRNA conc. (nM)

STOPS function optimally by interacting with multiple host factors

ALG-10000 lacks extensive base pairing

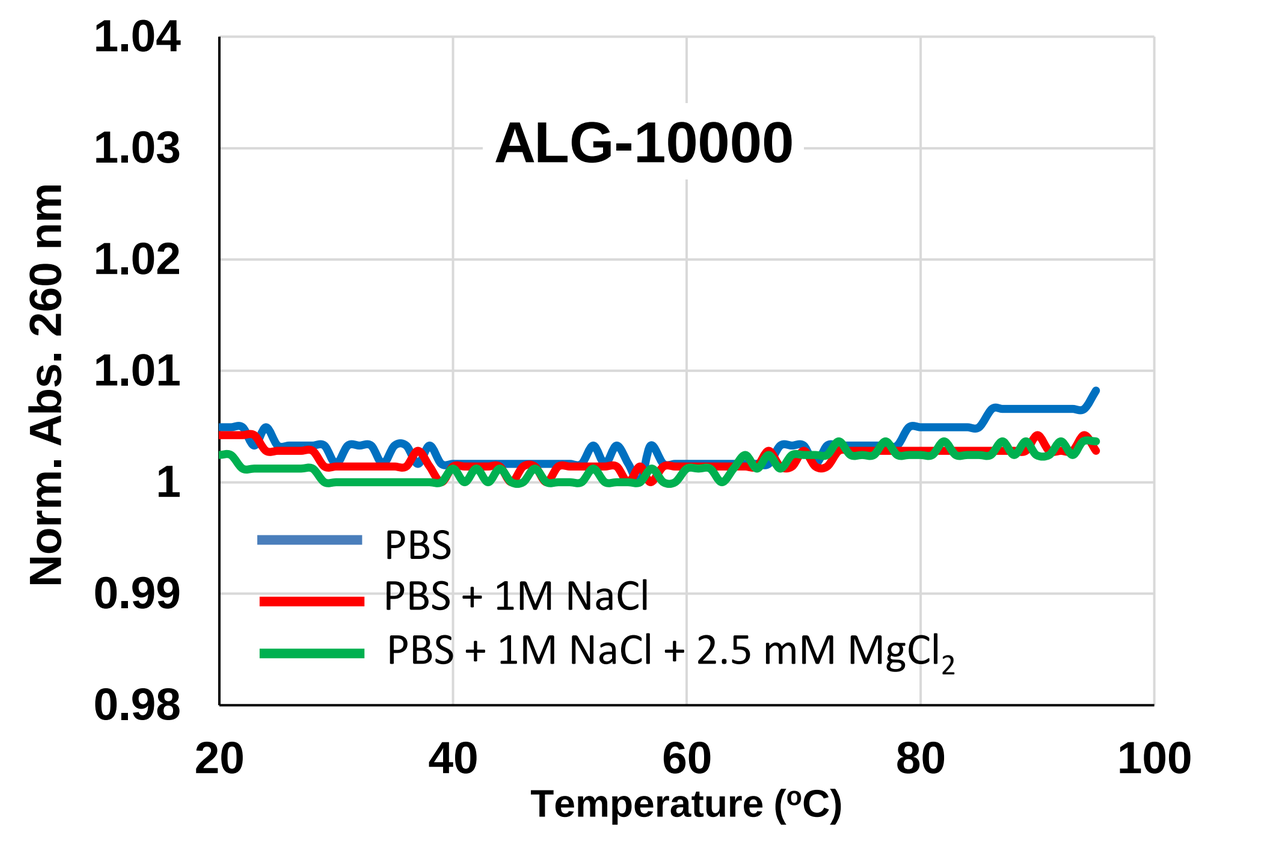

ALG-10000
Norm. Abs. 260 nm
PBS
PBS + 1M NaCl
PBS + 1M NaCl + 2.5 mM MgCl2
Temperature (°C)

STOPS base sequence affects interaction with HFs

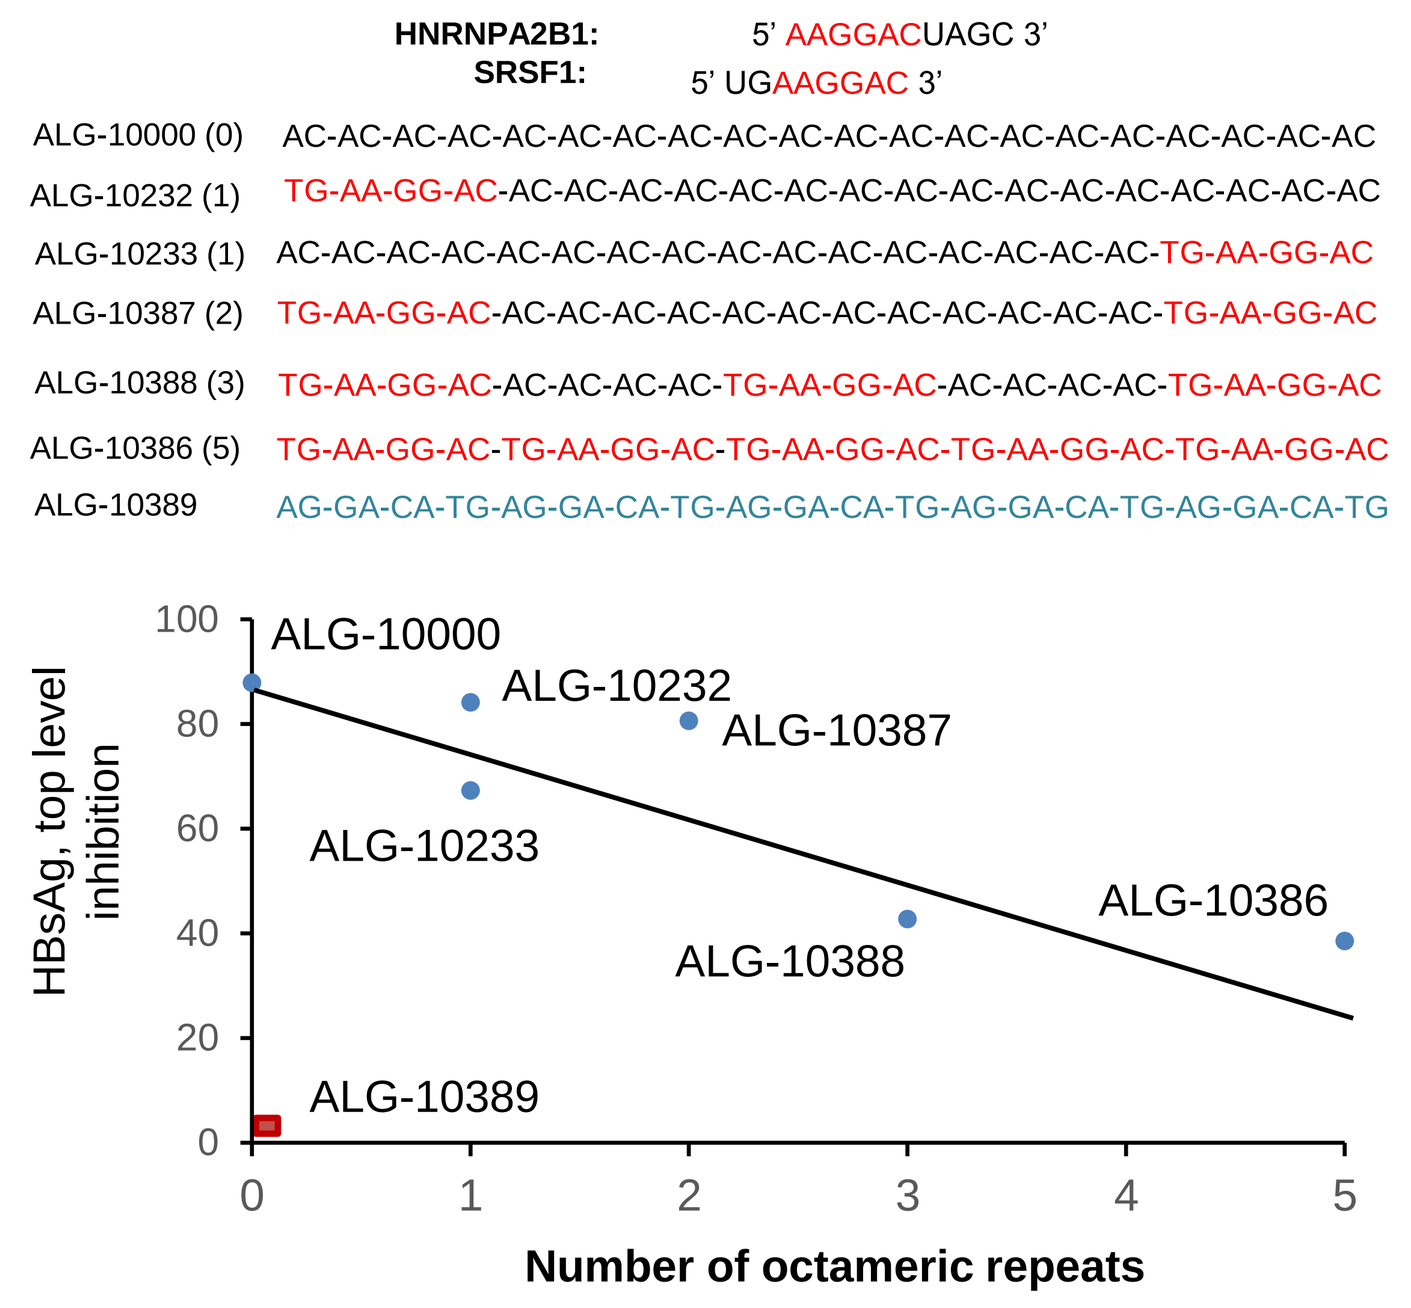

HNRNPA2B1: 5' AAGGACUAGC 3'
SRSF1: 5' UGAAGGAC 3'
ALG-10000 (0) AC-AC-AC-AC-AC-AC-AC-AC-AC-AC-AC-AC-AC-AC-AC-AC-AC-AC-AC-AC
ALG-10232 (1) TG-AA-GG-AC-AC-AC-AC-AC-AC-AC-AC-AC-AC-AC-AC-AC-AC-AC-AC-AC
ALG-10233 (1) AC-AC-AC-AC-AC-AC-AC-AC-AC-AC-AC-AC-AC-AC-AC-AC-TG-AA-GG-AC
ALG-10387 (2) TG-AA-GG-AC-AC-AC-AC-AC-AC-AC-AC-AC-AC-AC-AC-AC-TG-AA-GG-AC
ALG-10388 (3) TG-AA-GG-AC-AC-AC-AC-AC-TG-AA-GG-AC-AC-AC-AC-AC-TG-AA-GG-AC
ALG-10386 (5) TG-AA-GG-AC-TG-AA-GG-AC-TG-AA-GG-AC-TG-AA-GG-AC-TG-AA-GG-AC
ALG-10389 AG-GA-CA-TG-AG-GA-CA-TG-AG-GA-CA-TG-AG-GA-CA-TG-AG-GA-CA-TG
HBsAg, top level inhibition
ALG-10000
ALG-10232
ALG-10387
ALG-10233
ALG-10386
ALG-10388
ALG-10389
Number of octameric repeats

STOPS interaction with splicing factor cannot account for the complete HBsAg inhibition

The STOPS-binding host factors have different activities for HBV molecule production

HFs had major effects on:

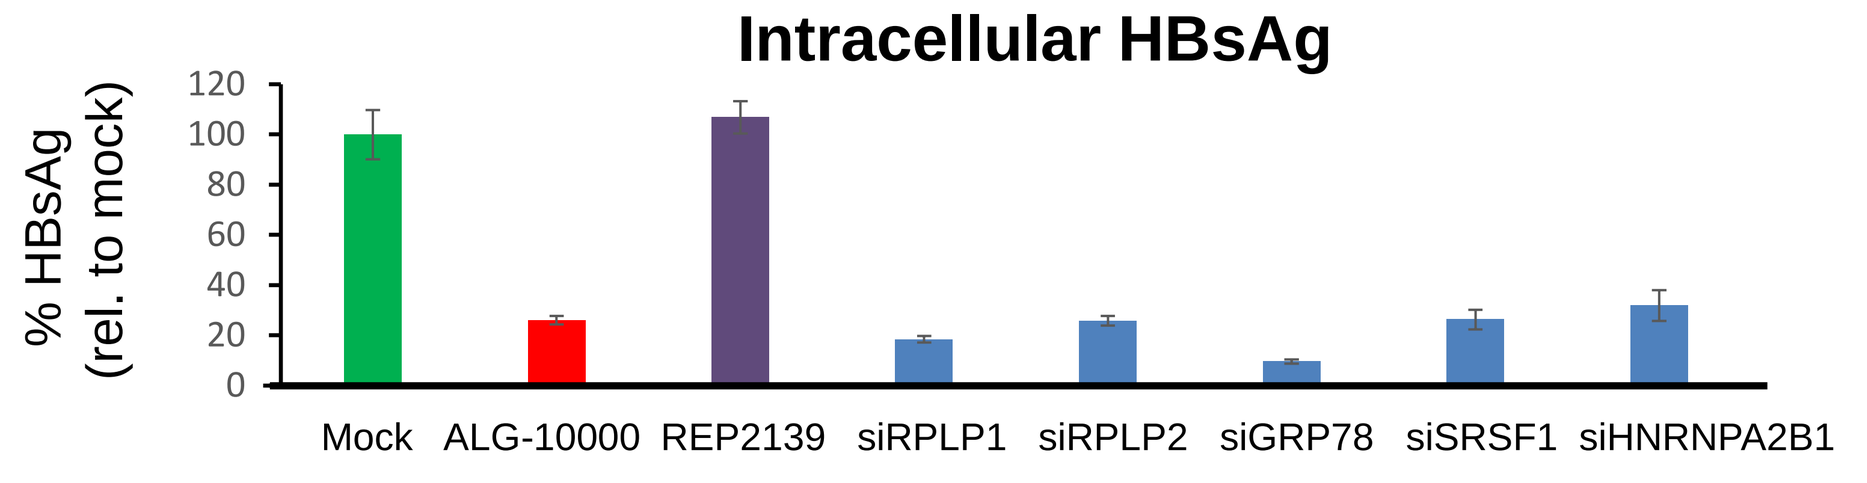

Intracellular HBsAg
% HBsAg (rel. to mock)
Mock
ALG-10000
REP2139
siRPLP1
siRPLP2
siGRP78
siSRSF1
siHNRNPA2B1

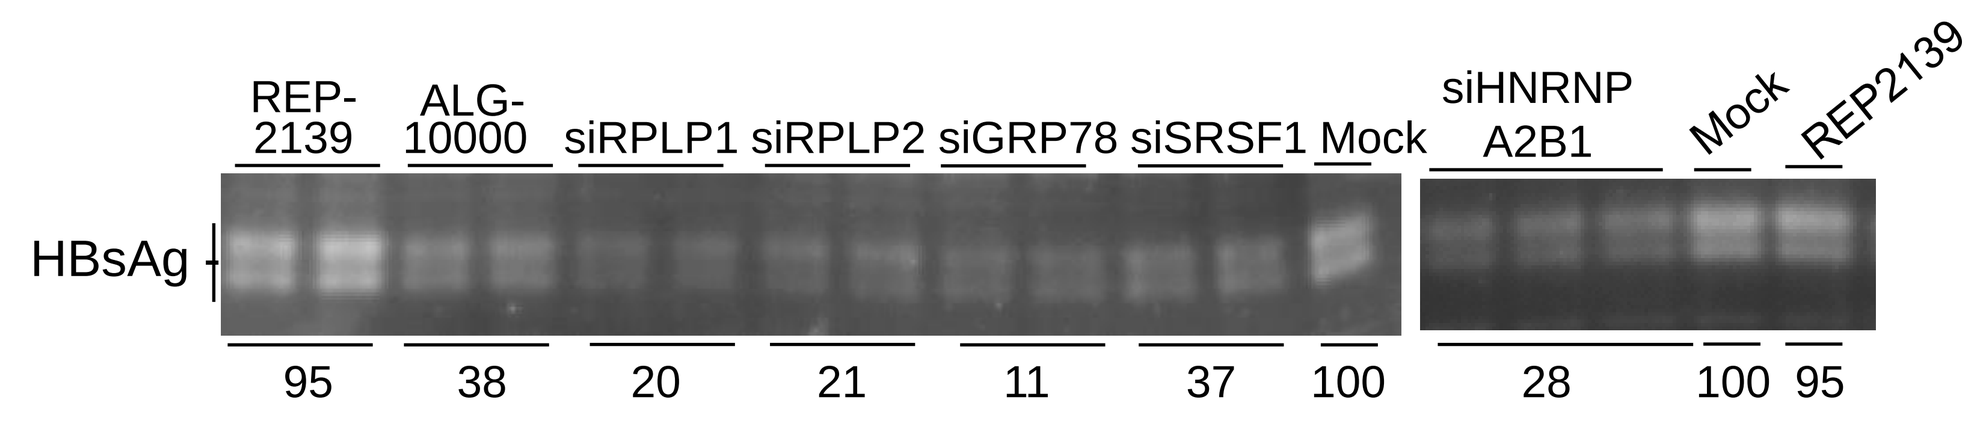

Intracellular HBsAg
REP-2139
ALG-10000
siRPLP1
siRPLP2
siGRP78
siSRSF1
Mock
siHNRNP A2B1
Mock
REP2139
HBsAg
95
38
20
21
11
37
100
28
100
95

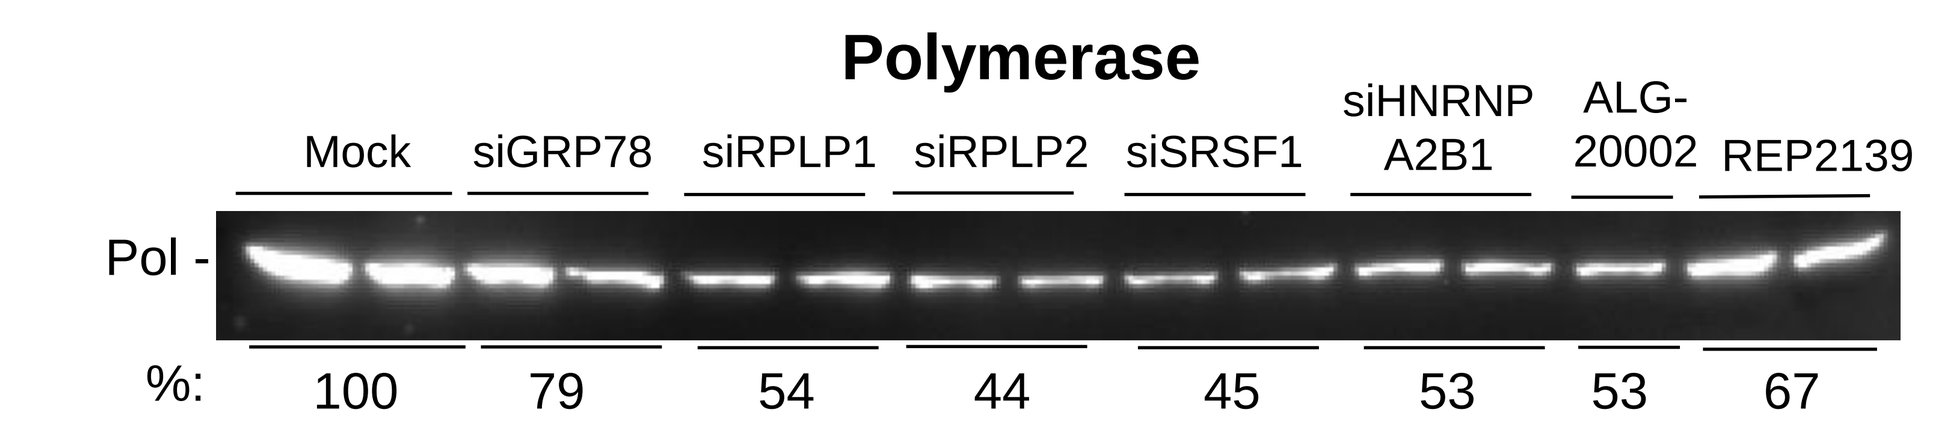

Polymerase
Mock
siGRP78
siRPLP1
siRPLP2
siSRSF1
siHNRNP A2B1
ALG-20002
REP2139
Pol -
%:
100
79
54
44
45
53
53
67

HFs had modest effects on:

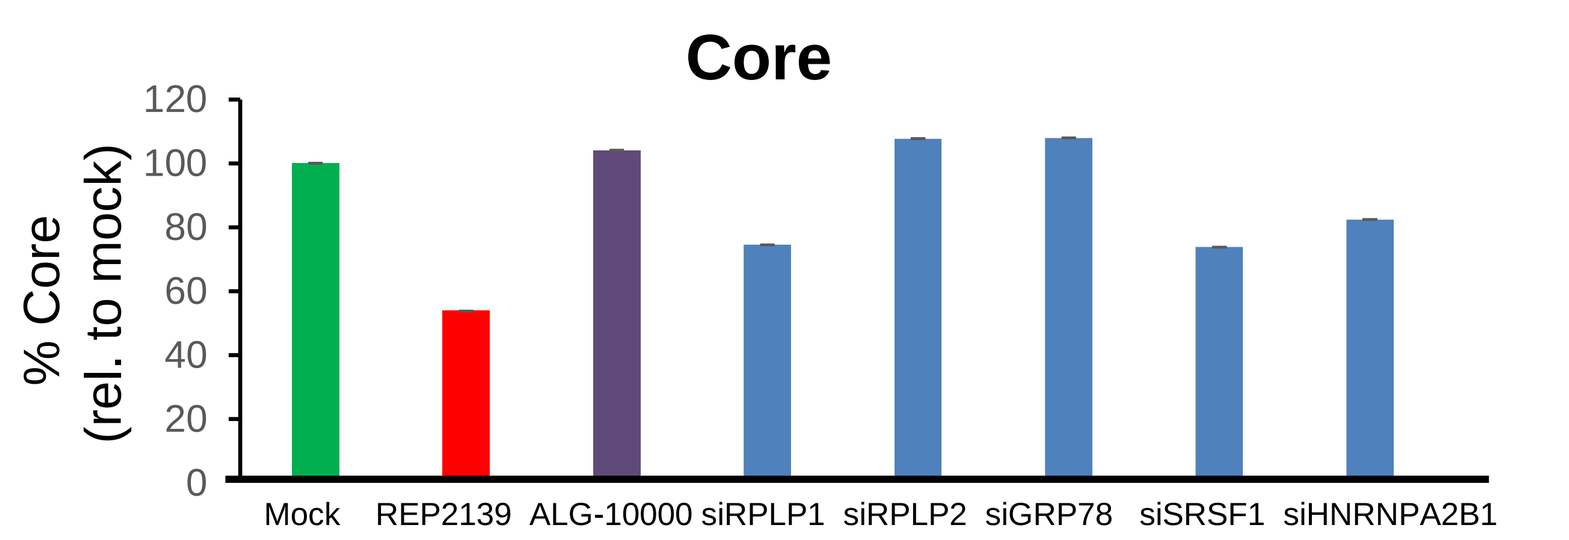

Core
% Core (rel. to mock)
Mock
REP2139
ALG-10000
siRPLP1
siRPLP2
siGRP78
siSRSF1
siHNRNPA2B1

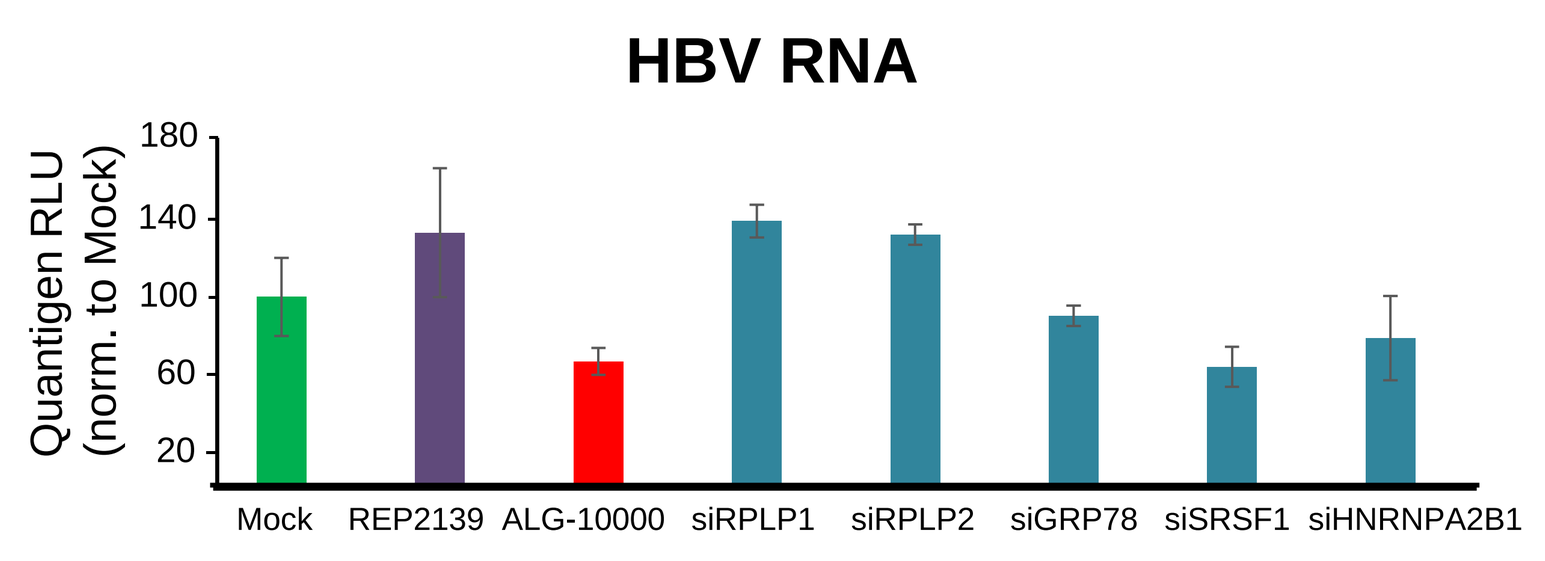

HBV RNA
Quantigen RLU (norm. to Mock)
Mock
REP2139
ALG-10000
siRPLP1
siRPLP2
siGRP78
siSRSF1
siHNRNPA2B1

STOPS and knockdowns of RPLP1/2 and GRP78 increase ubiquitination and proteasome degradation of HBsAg

IP for HBsAg and Western blot probed for Ub

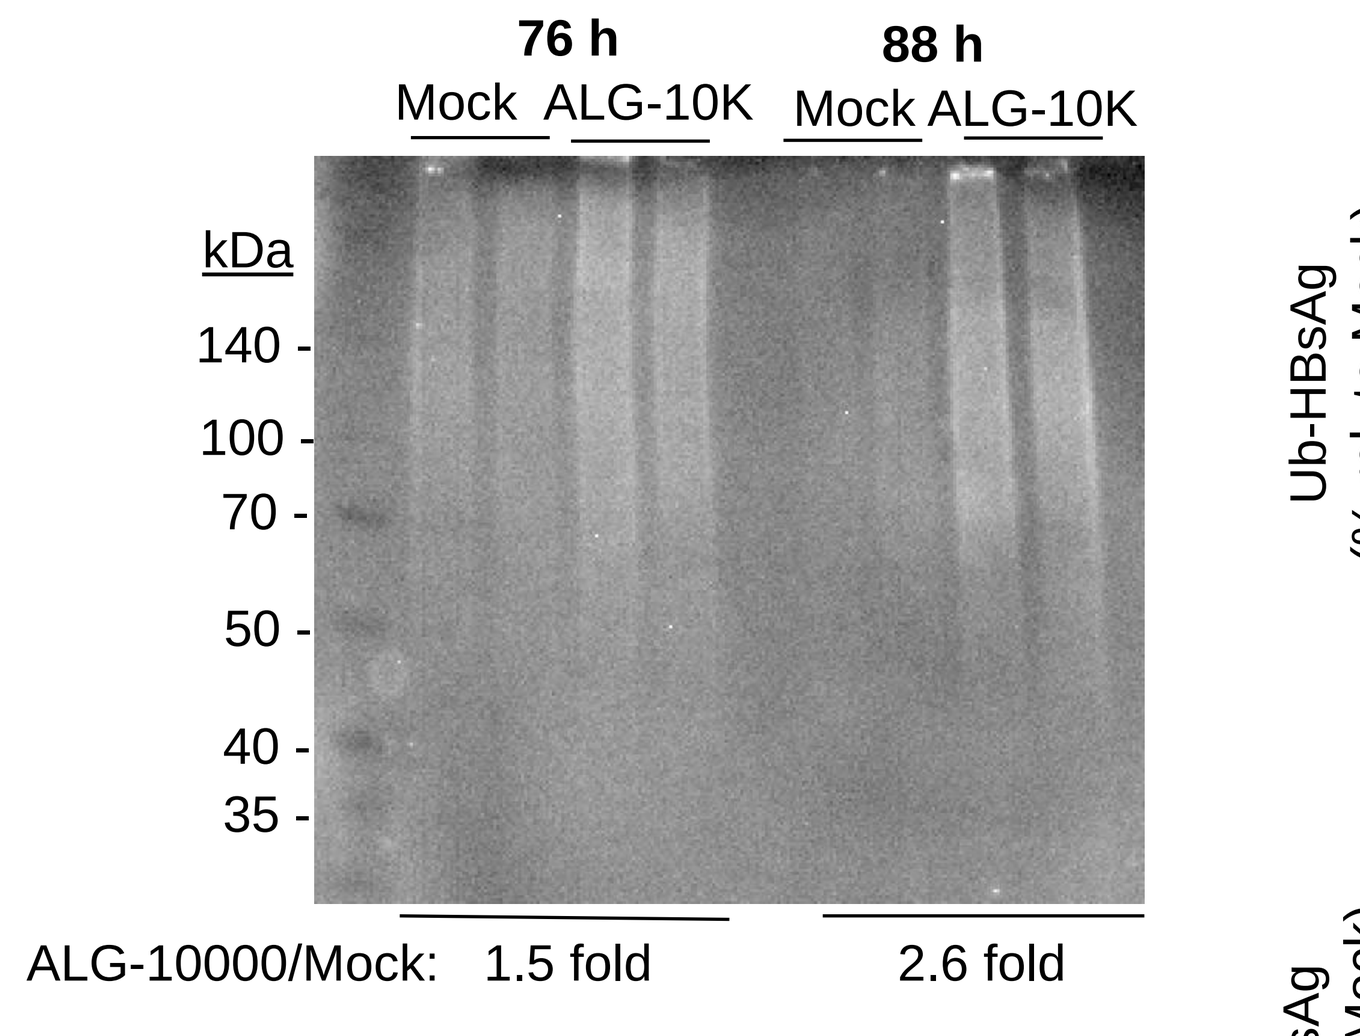

76 h
88 h
Mock
ALG-10K
Mock
ALG-10K
kDa
140
100
70
50
40
35
ALG-10000/Mock: 1.5 fold
2.6 fold

IP for Ub and Western blot probed for HBsAg

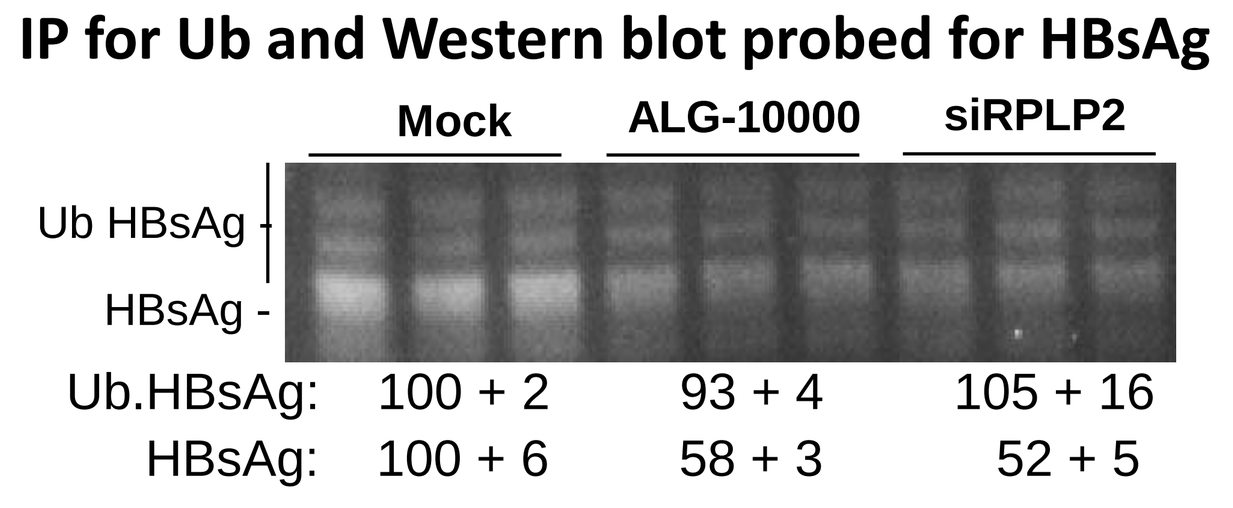

Mock
ALG-10000
siRPLP2
Ub HBsAg
HBsAg
Ub.HBsAg: 100 + 2 93 + 4 105 + 16
HBsAg: 100 + 6 58 + 3 52 + 5

ELISAs for Ub on HBsAg captured on plates

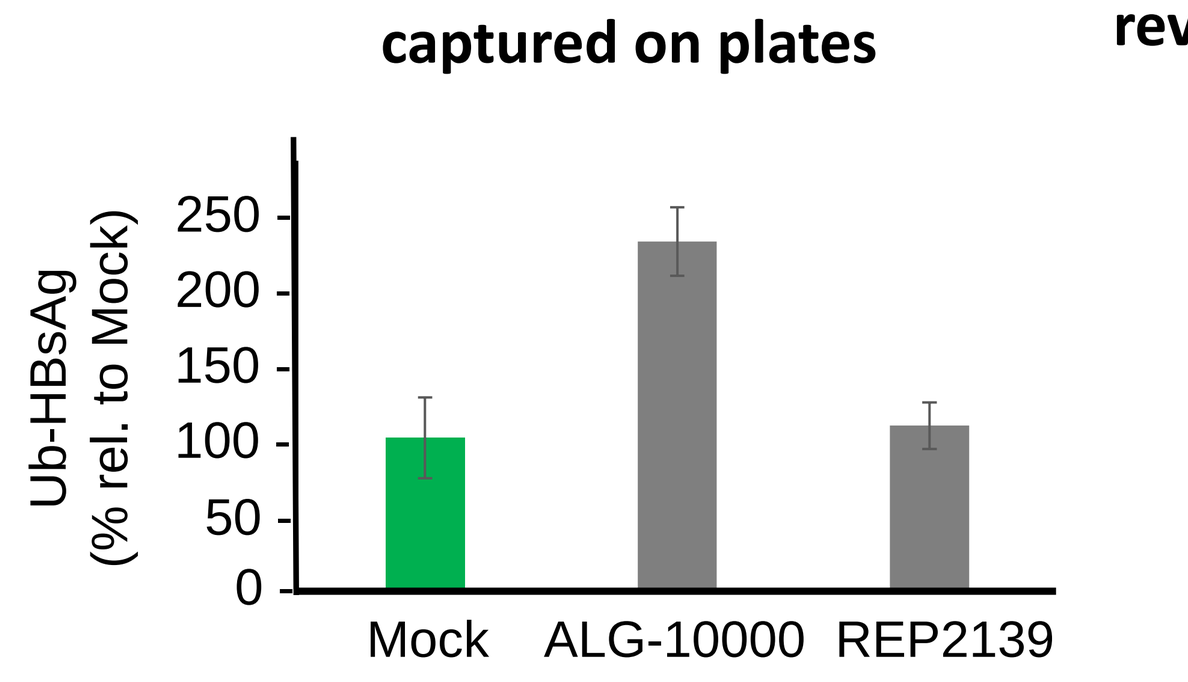

Ub-HBsAg (% rel. to Mock)
Mock
ALG-10000
REP2139

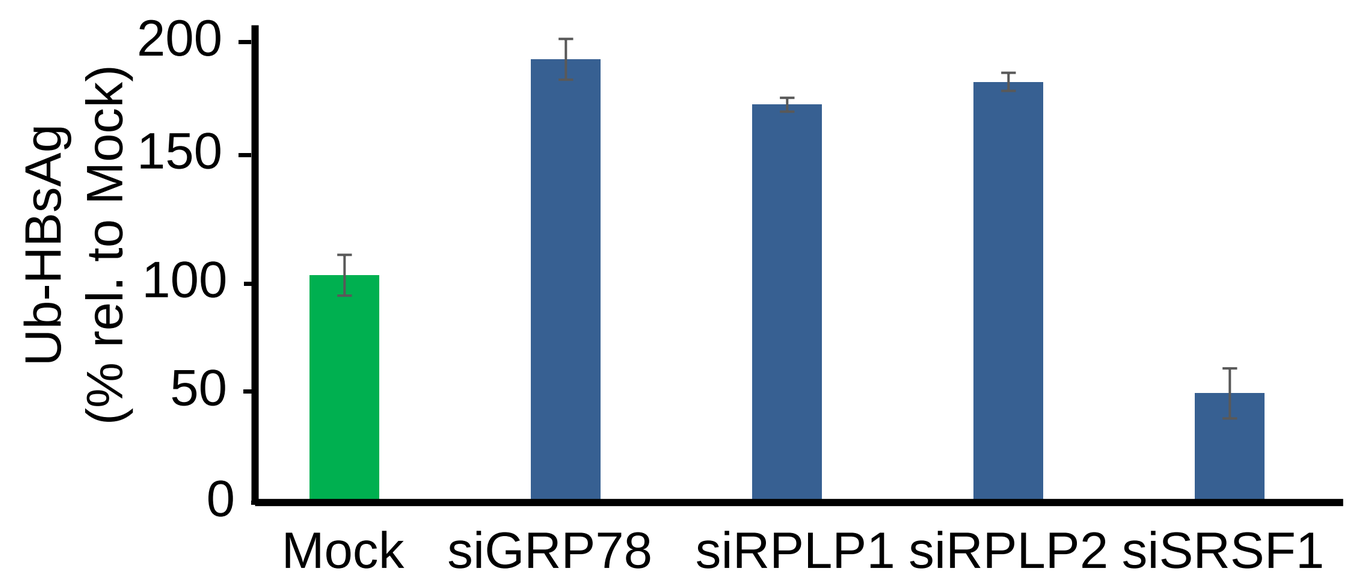

Ub-HBsAg (% rel. to Mock)
Mock
siGRP78
siRPLP1
siRPLP2
siSRSF1

Proteasome inhibitors can partially reverse the decrease in HBsAg abundance

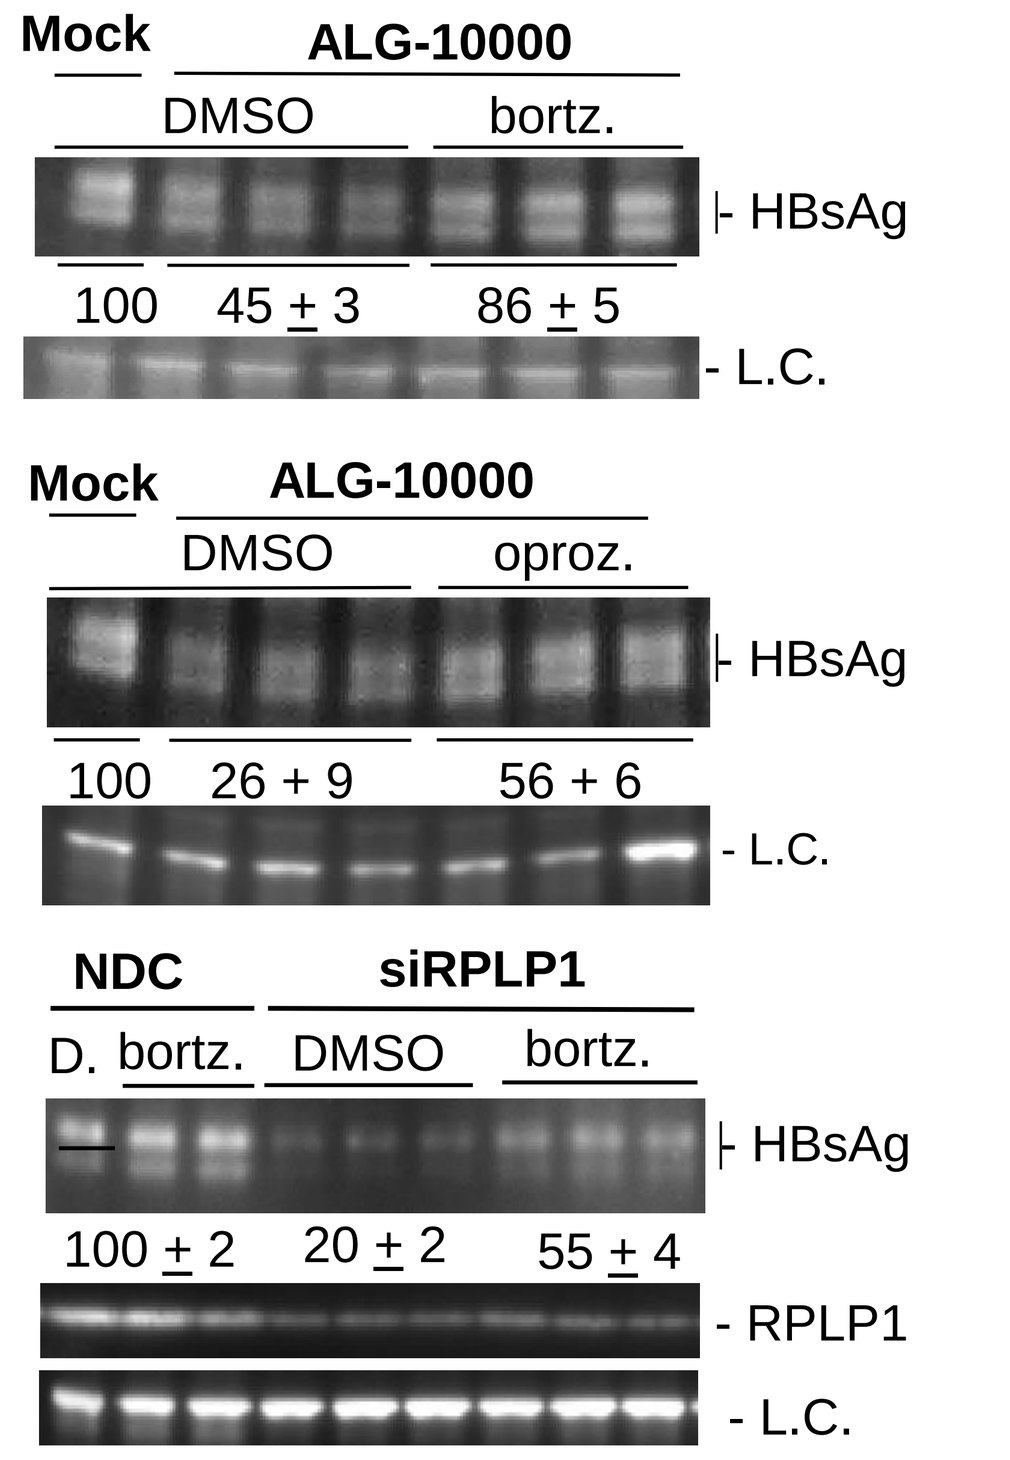

Mock
ALG-10000
DMSO
bortz.
HBsAg
100
45 ± 3
86 ± 5
L.C.
Mock
ALG-10000
DMSO
oproz.
HBsAg
100
26 + 9
56 + 6
L.C.
NDC
siRPLP1
D.
bortz.
DMSO
bortz.
HBsAg
100 ± 2
20 ± 2
55 ± 4
RPLP1
L.C.

Conclusions and Discussion

STOPS have a complex mechanism of action to inhibit HBV infection and HBsAg production. Through sequestration of cellular factors involved in RNA processing, STOPS decrease the expression of HBV molecules. Through the sequestration of cellular factors needed to translate and properly fold the HBV S-antigen, increase their ubiquitination and intracellular degradation.